Evaluation of a Novel Machine Learning Method for PARP Inhibitor Sensitivity Prediction Using Real-World Data

ZEPHYR AI

Emily Vucic, Felicia Kuperwaser, Sunil Kumar, Dillon Tracy, Olena Marchenko, Zong Miao, Jacob Kaffey, Jordan Wolinsky, Sean Klei, Andrey Chursov, Fahad Khan, Jean Michel Rouly, George Komatsoulis, Jeff Sherman, Rachael Brake and Maayan Baron

Zephyr AI: 1800 Tysons Blvd, Suite 901
McLean VA 22102 | www.zephyrai.bio

Methods to Stratify Late-Line Cancer Patients Responsive to PARP Inhibitors is an Unmet Need

- Tumors with homologous recombination repair deficiencies (HRD+) are susceptible to PARP inhibitors (PARPi) which exploit a synthetic lethality by targeting an essential base excision repair (BER) pathway. Four drugs are approved for use with this biomarker.
- However, current patient selection biomarkers designed to enrich for PARPi response (HRD+, BRCA1/2^{mut}) in late-line cancer patients are insufficient. This has led to withdrawals of these drugs in certain indications, including late-line ovarian cancer.
- We aimed to develop and validate an alternative, artificial intelligence /machine learning (AI/ML) based method to identify olaparib sensitive ovarian cancer patients

The Zephyr Model Harnesses Insights From Vast, Diverse Multi-Modal Datasets to Inform & Enhance Decision-Making

- The Zephyr foundation model transforms clinical, translational and chemical data into rich 'machine learning-ready' representations using custom encoders. These data representations are then integrated into a neural network forming Zephyr's foundation drug response & Vulnerability Network™ prediction model (**Figure 1, center**).

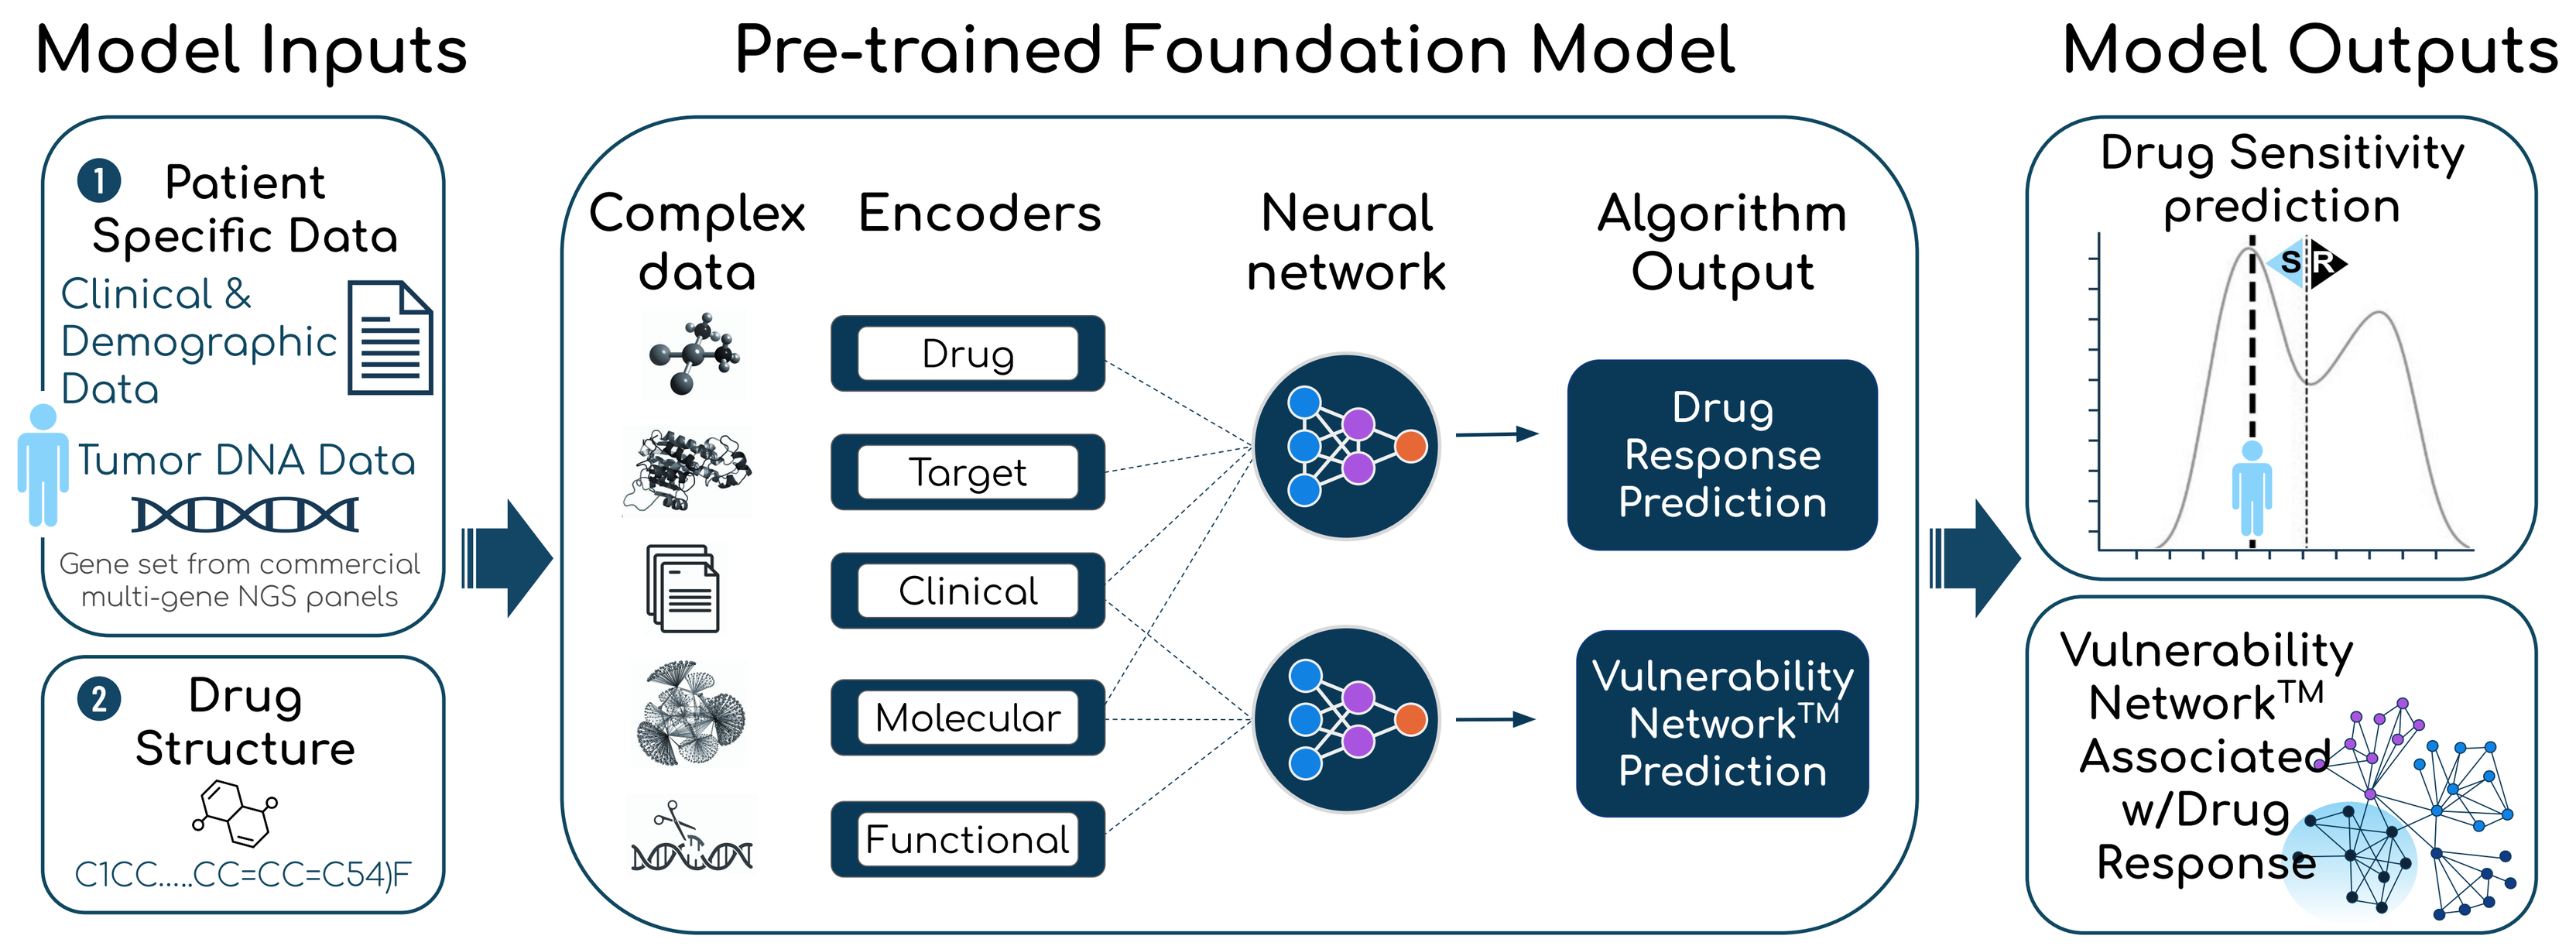
Model Inputs
Pre-trained Foundation Model
Model Outputs
1 Patient Specific Data
Clinical & Demographic Data
Tumor DNA Data
Gene set from commercial multi-gene NGS panels
2 Drug Structure
C1CC.....CC=CC=C54)F
Complex data
Encoders
Neural network
Algorithm Output
Drug
Target
Clinical
Molecular
Functional
Drug Response Prediction
Vulnerability Network™ Prediction
Drug Sensitivity prediction
Vulnerability Network™ Associated w/Drug Response

Figure 1 | Overview of Zephyr's AI/ML Foundation Model for Generating Biologically Interpretable Drug Response Predictions in Cancer Patients Using Complex Clinically Relevant Data.

- Interpretable drug response predictions for individual cancer patients can be generated by using the Zephyr foundation model and patient-specific input data. Namely, clinico-genomic data and a structural embedding of the drug of interest (**left panel**).
- For each prediction, two model outputs are generated (**right panel**): a drug sensitivity prediction for a specific drug and a Vulnerability Network™ (VN) representing predicted perturbation sensitivities operative in a tumor. Drug response sensitivity thresholds are set based on predictions from large real-world patient cohorts.
- For drug combination predictions, each drug in the combination is run through the model individually. A predicted response to the combination per patient, is made based on concordant predictions for each drug in the combination (e.g., predicted sensitive to all drugs in a combination is classified as 'sensitive' for that combination).

Baseline Characteristics of Real-World Olaparib-Treated Ovarian Cancer Patients for Zephyr Response Prediction Validation

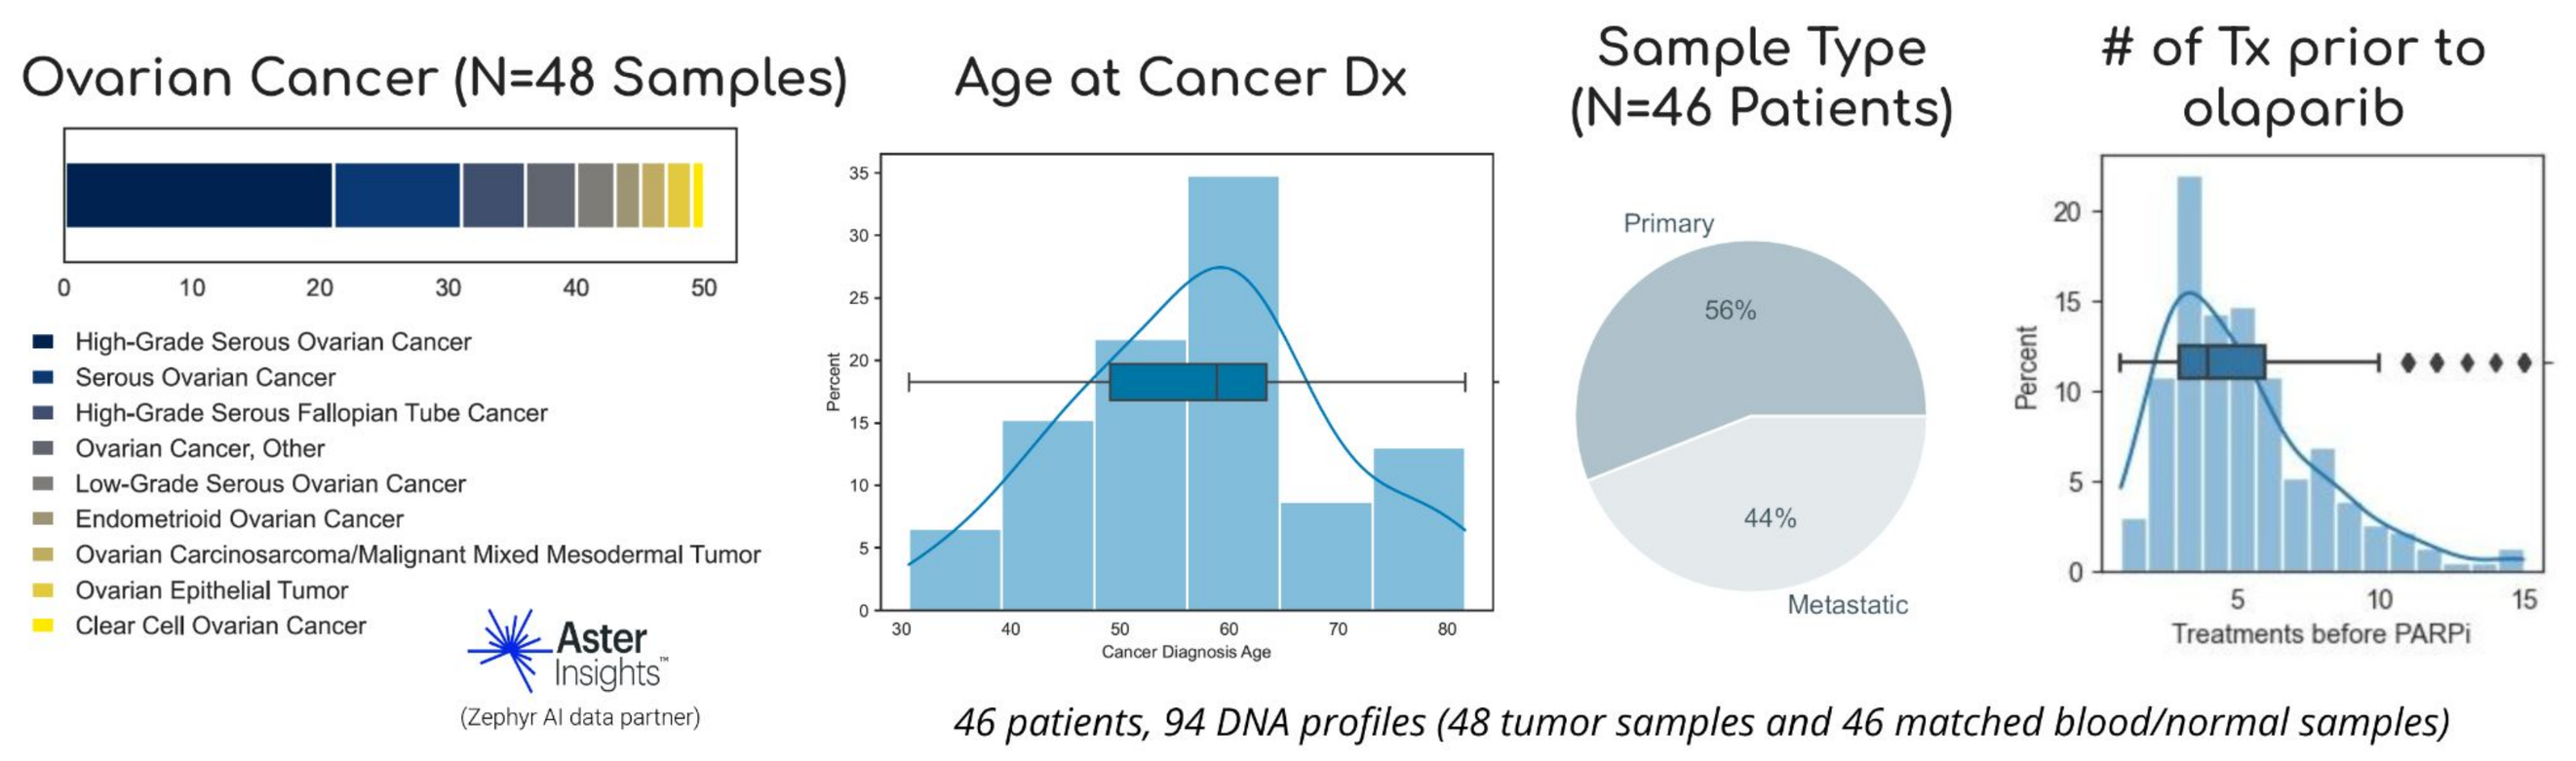
Ovarian Cancer (N=48 Samples)
High-Grade Serous Ovarian Cancer
Serous Ovarian Cancer
High-Grade Serous Fallopian Tube Cancer
Ovarian Cancer, Other
Low-Grade Serous Ovarian Cancer
Endometrioid Ovarian Cancer
Ovarian Carcinosarcoma/Malignant Mixed Mesodermal Tumor
Ovarian Epithelial Tumor
Clear Cell Ovarian Cancer
Aster Insights
(Zephyr AI data partner)
Age at Cancer Dx
Cancer Diagnosis Age
Sample Type (N=46 Patients)
Primary 56%
Metastatic 44%
of Tx prior to olaparib
Treatments before PARPi
46 patients, 94 DNA profiles (48 tumor samples and 46 matched blood/normal samples)

Figure 2 | Characteristics of olaparib treated ovarian cancer validation cohort. Input data required for drug response predictions were obtained for 46 ovarian cancer patients. Treatment histories for these patients were complex, with diverse drug combinations and a variable number of prior lines of treatment prior to receiving olaparib. Samples were restricted to NGS data from biopsies collected no more than two years prior to the initiation of olaparib, with the aim of minimizing the impact of clonal evolution, genetic drift, and selective pressures from intervening treatments on the relevance of the Zephyr model drug response predictions for olaparib.

Zephyr's Method Identifies Ovarian Cancer Patients with Improved Real-World (rw) Outcomes Over Conventional Stratification Approaches

- Conventional HRD+ stratification for olaparib sensitivity was contrasted with the use of the Zephyr AI/ML model predictions. Patient sensitivity outcomes were represented as real world-overall survival (rw-OS) or rw-progression-free survival (rw-PFS).
- Conventional HRD+ stratification did not result in statistically significant improvement in either of the rw outcome whereas, for the same cohort, patient stratification via the Zephyr model significantly improved outcomes for both real world end points. (**Compare Figure 3A vs. 3B**).
- Crucially, these results were unaffected by BRCA1/2 mutations, HRD+ status, or other clinical confounders (**Figure 3C**), underscoring that the olaparib-sensitive patients identified by Zephyr's method cannot be detected through conventional HRD+ stratification methods.

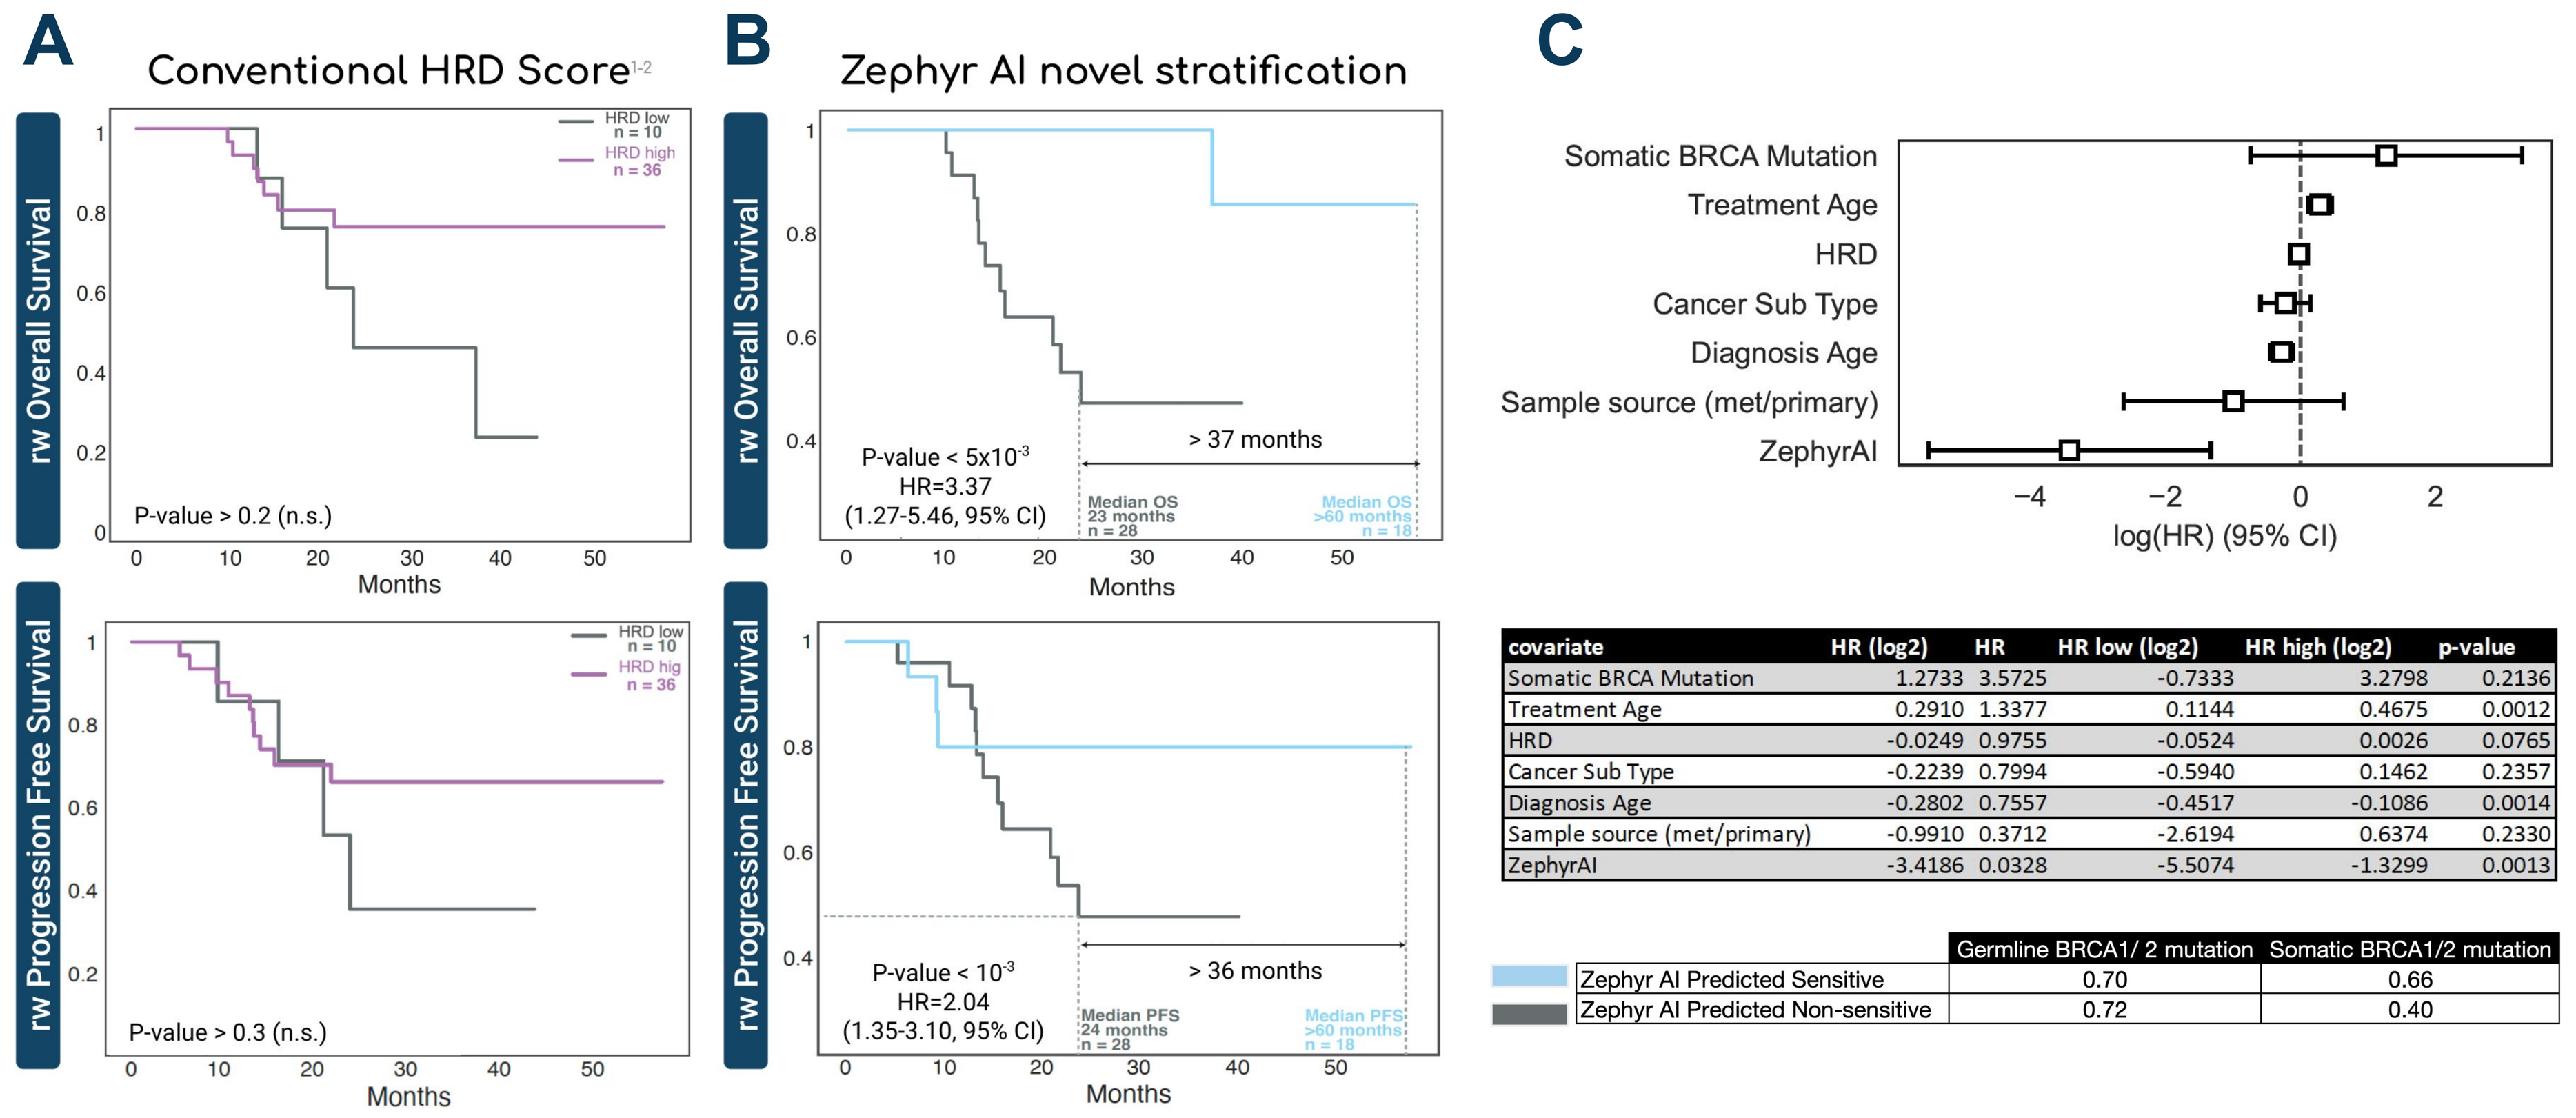
A
Conventional HRD Score1-2
rw Overall Survival
HRD low n = 10
HRD high n = 36
P-value > 0.2 (n.s.)
Months
rw Progression Free Survival
P-value > 0.3 (n.s.)
B
Zephyr AI novel stratification
P-value < 5x10-3
HR=3.37
(1.27-5.46, 95% CI)
> 37 months
Median OS 23 months n = 28
Median OS >60 months n = 18
P-value < 10-3
HR=2.04
(1.35-3.10, 95% CI)
> 36 months
Median PFS 24 months n = 28
Median PFS >60 months n = 18
C
Somatic BRCA Mutation
Treatment Age
HRD
Cancer Sub Type
Diagnosis Age
Sample source (met/primary)
ZephyrAI
log(HR) (95% CI)

covariate	HR (log2)	HR	HR low (log2)	HR high (log2)	p-value
Somatic BRCA Mutation	1.2733	3.5725	-0.7333	3.2798	0.2136
Treatment Age	0.2910	1.3377	0.1144	0.4675	0.0012
HRD	-0.0249	0.9755	-0.0524	0.0026	0.0765
Cancer Sub Type	-0.2239	0.7994	-0.5940	0.1462	0.2357
Diagnosis Age	-0.2802	0.7557	-0.4517	-0.1086	0.0014
Sample source (met/primary)	-0.9910	0.3712	-2.6194	0.6374	0.2330
ZephyrAI	-3.4186	0.0328	-5.5074	-1.3299	0.0013

	Germline BRCA1/ 2 mutation	Somatic BRCA1/2 mutation
Zephyr AI Predicted Sensitive	0.70	0.66
Zephyr AI Predicted Non-sensitive	0.72	0.40

Figure 3 | Evaluation of Zephyr Olaparib Response Predictions Using Real-World Ovarian Cancer Patient Outcomes. 'Conventional HRD' scores were derived from Whole Exome Sequencing (WES) data, using methods that assess Allelic Imbalance (AI), somatic and germline hits to HRR genes, and an HRD threshold of ≥42. **A)** Stratification of ovarian cancer patients treated with olaparib by conventional HRD scores does not result in significant differences in rw-overall (rw-OS) (**top**) or progression free (rw-PFS) (**bottom**) survival. **B)** In contrast, stratification of the same patients by Zephyr's method results in statistically and clinically significant prolonged rw-OS (**top**) and rw-PFS (**bottom**). Response predictions were not confounded by BRCA1/2 somatic or germline mutations, HRD status or assessed potential clinical confounders (**C**).

Evidence of Intact DNA Damage Response Pathways in Zephyr-Predicted Olaparib Insensitive Ovarian Cancer Tumors

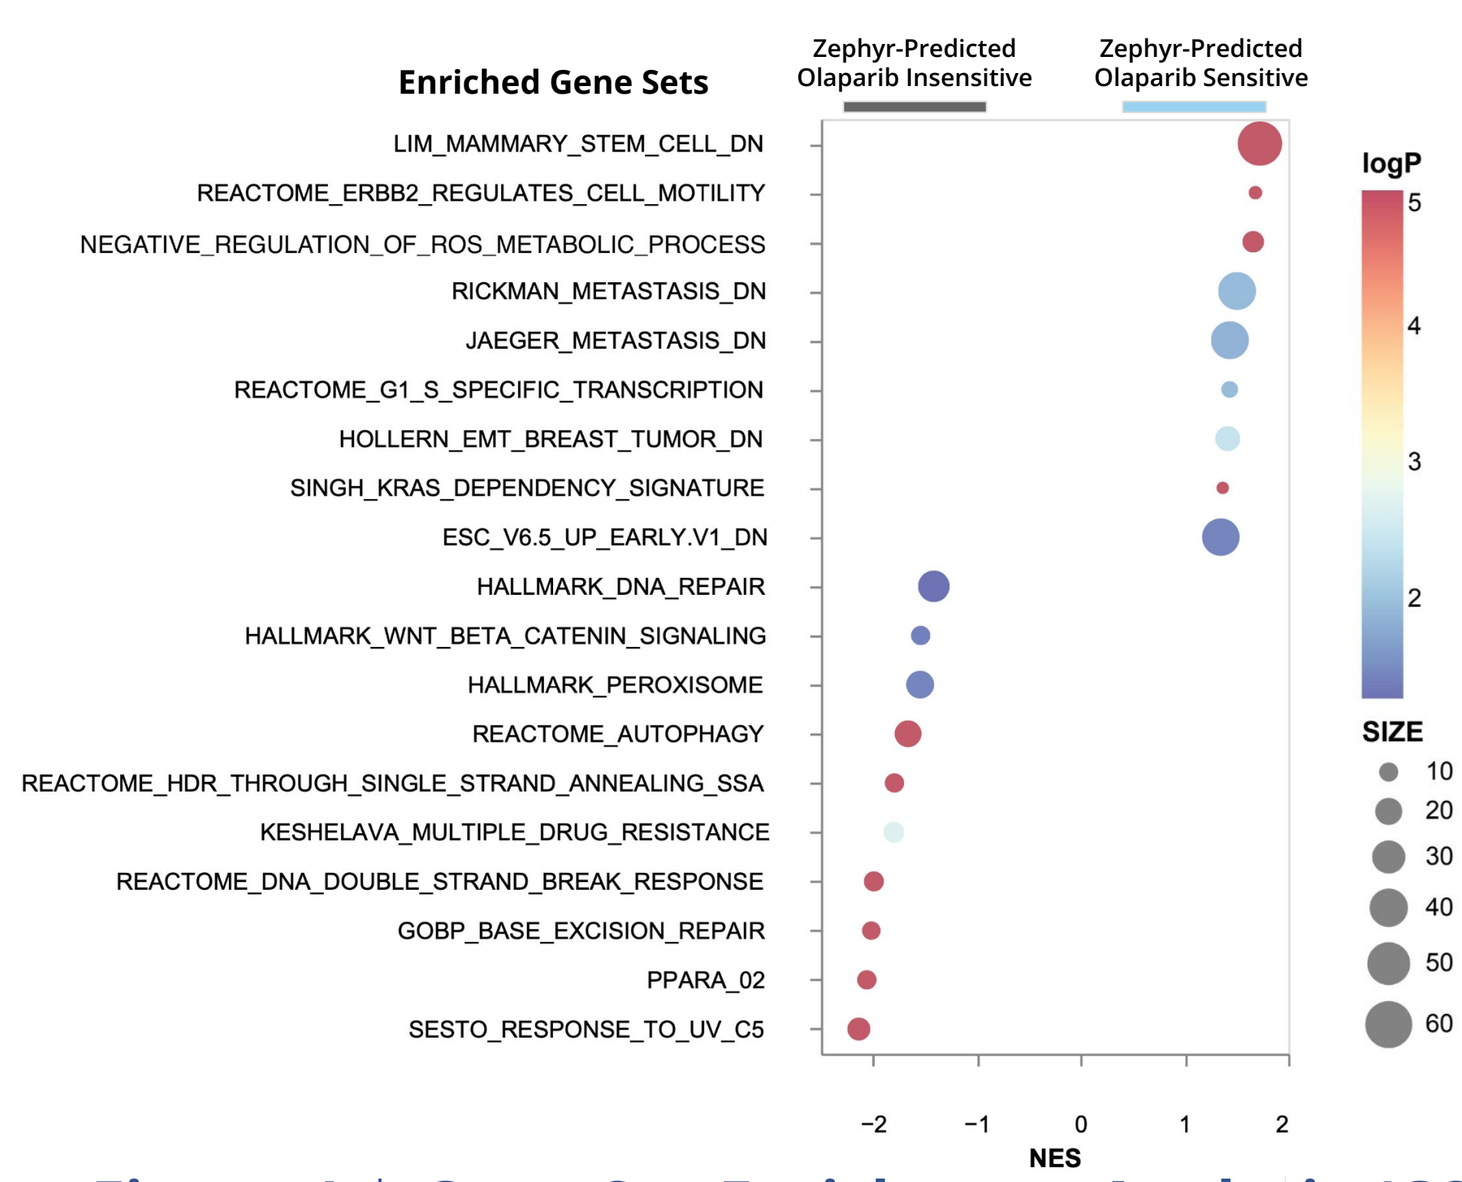
Enriched Gene Sets
Zephyr-Predicted Olaparib Insensitive
Zephyr-Predicted Olaparib Sensitive
LIM_MAMMARY_STEM_CELL_DN
REACTOME_ERBB2_REGULATES_CELL_MOTILITY
NEGATIVE_REGULATION_OF_ROS_METABOLIC_PROCESS
RICKMAN_METASTASIS_DN
JAEGER_METASTASIS_DN
REACTOME_G1_S_SPECIFIC_TRANSCRIPTION
HOLLERN_EMT_BREAST_TUMOR_DN
SINGH_KRAS_DEPENDENCY_SIGNATURE
ESC_V6.5_UP_EARLY.V1_DN
HALLMARK_DNA_REPAIR
HALLMARK_WNT_BETA_CATENIN_SIGNALING
HALLMARK_PEROXISOME
REACTOME_AUTOPHAGY
REACTOME_HDR_THROUGH_SINGLE_STRAND_ANNEALING_SSA
KESHELAVA_MULTIPLE_DRUG_RESISTANCE
REACTOME_DNA_DOUBLE_STRAND_BREAK_RESPONSE
GOBP_BASE_EXCISION_REPAIR
PPARA_02
SESTO_RESPONSE_TO_UV_C5
logP
SIZE
NES

- Reconstructed mRNA profiles[3] for ovarian patient tumors were used as input for GSEA comparing Zephyr predicted olaparib sensitive vs. insensitive tumors.
- Predicted-sensitive tumors were positively enriched for proliferation signatures and negatively enriched for metastatic, stem-like, and EMT signatures.
- Predicted-insensitive tumors were positively enriched for multiple DNA damage response pathways, suggesting that despite presence of BRCA1/2 mutations and HRD+ status these pathways remain functional in these tumors, potentially bypassing the synthetic lethality mechanism exploited by PARP inhibitors.

Figure 4 | Gene Set Enrichment Analysis (GSEA) in Zephyr Olaparib Predicted Insensitive vs. Sensitive Ovarian Cancer Tumors Using Reconstructed mRNA. Normalized enrichment scores (NES) (x-axis) indicate enrichment of corresponding gene sets (y axis) in Zephyr predicted olaparib-sensitive tumors (NES ≥ 1) or Zephyr predicted olaparib non-sensitive tumors (NES ≤ -1). Circle size indicates number of genes per set; color denotes log-transformed p-value.

Tumors Predicted to Be Sensitive to Olaparib Reveal Dependencies on Homologous Recombination and Cell Cycle Checkpoint Machinery

Vulnerability Networks™ (VNs) represent predicted perturbation sensitivities operative in tumors. Zephyr-predicted olaparib-sensitive tumors are characterized by VNs enriched for HR machinery, suggesting a reliance on this pathway in these tumors that may explain PARP inhibitor sensitivity (**Figure 5A**). Conversely, as supported by GSEA (**Figure 4**), VNs enriched in insensitive tumors show no predicted susceptibility to HR pathway perturbation.

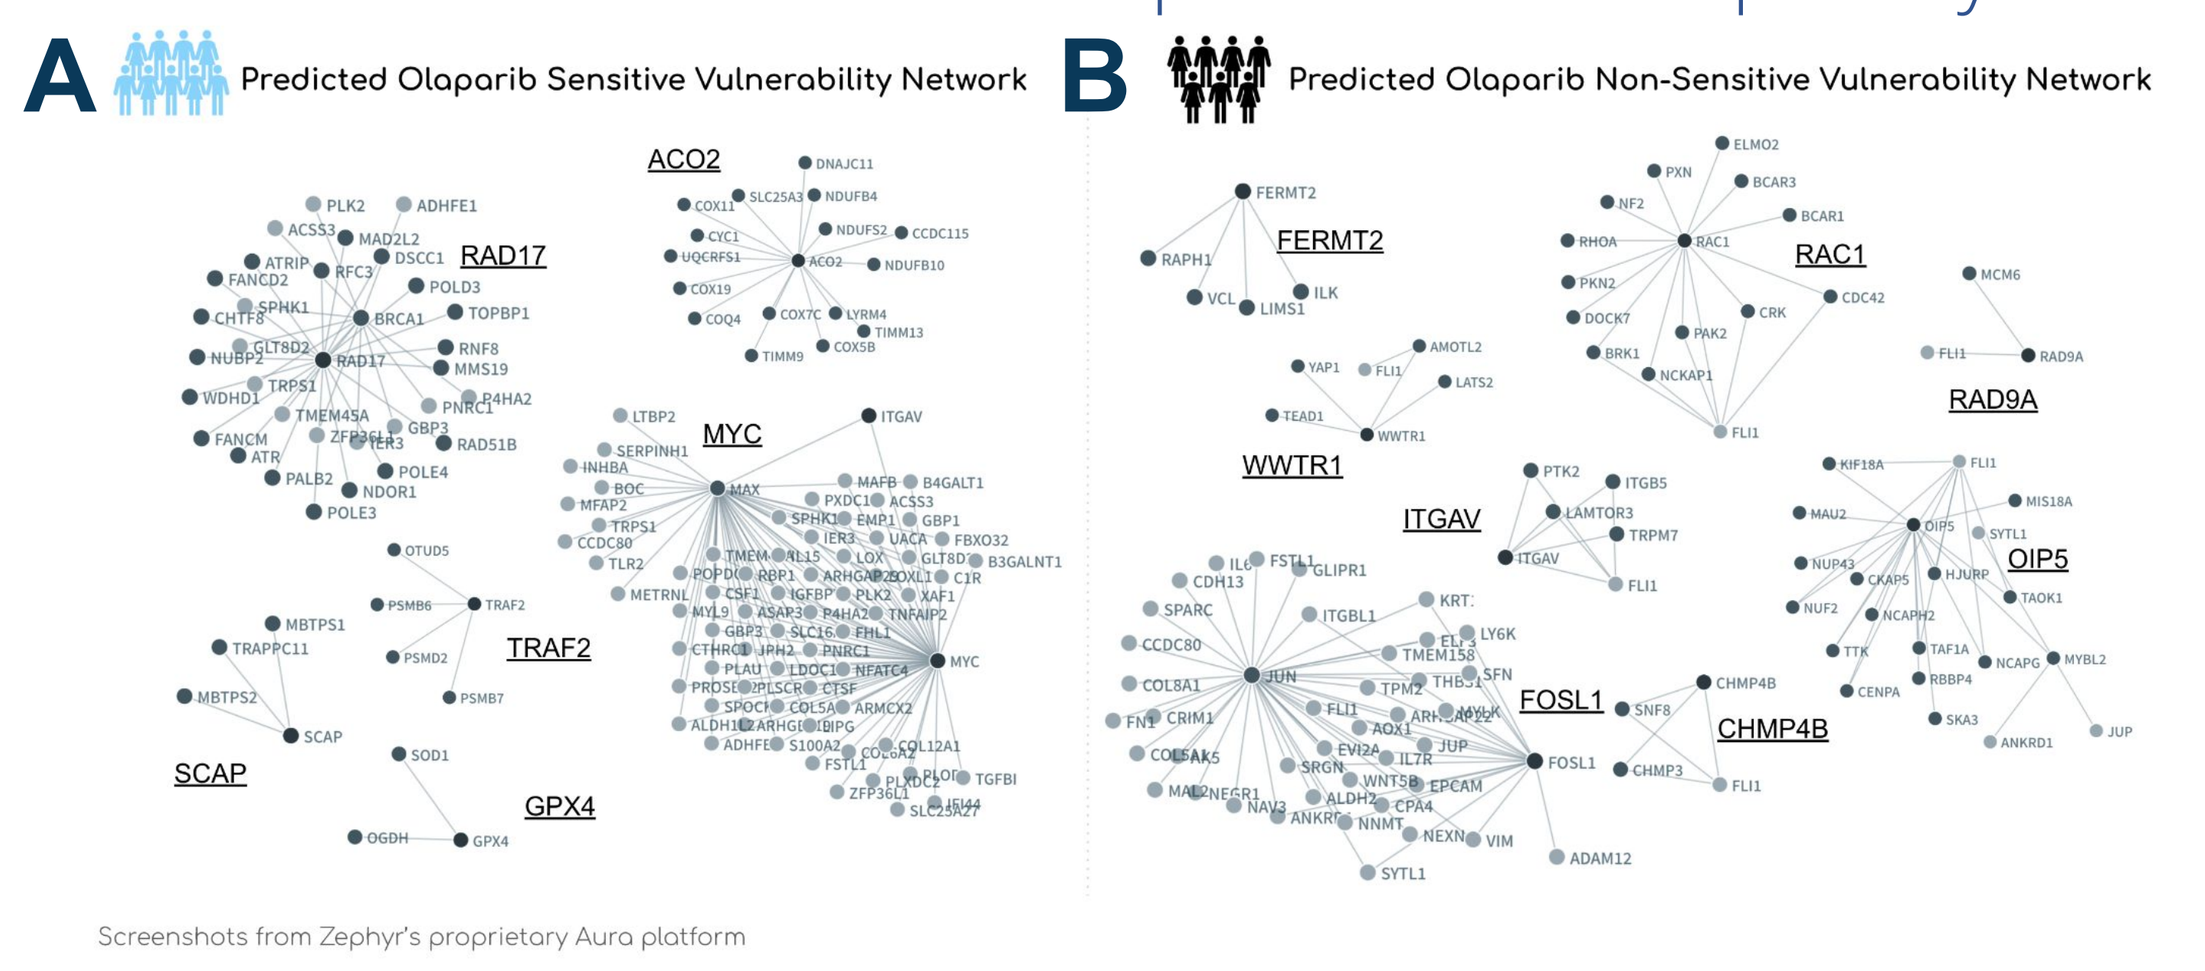
A Predicted Olaparib Sensitive Vulnerability Network
B Predicted Olaparib Non-Sensitive Vulnerability Network
ACO2
RAD17
MYC
TRAF2
SCAP
GPX4
FERMT2
RAC1
RAD9A
WWTR1
ITGAV
OIP5
FOSL1
CHMP4B
Screenshots from Zephyr's proprietary Aura platform

Instead, these tumors are characterized by a predicted dependence on cell motility and stress response mechanisms, which could potentially bypass PARP inhibitor effects and contribute to the poor olaparib response observed in these patients (**Figure 5B**).

Figure 5 | Vulnerability Networks™ (VNs) Enriched in Zephyr Predicted Olaparib Sensitive and Insensitive Ovarian Tumors. VNs depict predicted perturbation sensitivities our models identify as operative in tumors, potentially explaining drug responses. Nodes represent predicted perturbation sensitivities. Edges are formed based on established and inferred protein-protein and protein-DNA interactions. Underlined genes represent subcluster network hubs.

Our Novel AI/ML Method Identifies Patients Likely to Respond to Olaparib, Yielding Enhanced rw-Outcomes Across Late-Stage Ovarian Cancer & Complex Combination Treatment Contexts Independent of HRD status

- Zephyr has developed an AI/ML method that integrates and harnesses insights from large, diverse and disparate multi-modal datasets, to create a novel patient selection tool that uniquely advances precision medicine.
- In this study, we used a real-world ovarian cancer cohort to validate our model's ability to identify olaparib-sensitive patients for whom existing biomarkers are insufficient, addressing a critical unmet need in precision oncology.
- Using RWD, our models are conducive to both prospective and retrospective validation.

REFERENCES: **[1]** Rempel et al. NPJ Precis Oncol. 2022 Jun 9;6(1):36; **[2]** Vogel et al. Diagn Pathol. 2024 Jan 6;19(1):9; GATK4, sequenza; scarHRD; **[3]** Kumar S et al. Reconstructing a latent representation of gene expression from genomic alterations to improve clinical utility of real-world clinicogenomics data. American Association for Cancer Research 2024 Annual Meeting. Abstract # 3519

ACKNOWLEDGEMENTS: The authors express their gratitude to the Zephyr AI science, engineering, data and business development teams for invaluable technical support and discussion, especially Candy Zhu and Jasmine Chu for their valuable assistance in designing this poster.

Copies of this poster/slide deck obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® or the authors of this poster/slides.