Identifying Novel Drivers of Drug Sensitivity using an AI-Enabled Multi-Modal Biomarker – Osimertinib sensitivity beyond EGFR

ZEPHYR AI

Maayan Baron, Felicia Kuperwaser, Colin Tang, Sunil Kumar, Dillon Tracy, Zong Miao, Sepideh Foroutan, Amy Sheide, Nathaniel Tann, Samantha Pindak, Jacob Kaffey, Jordan Wolinsky, Sean Klei, Brandon Funkhouser, Nick Lee, Patrick Bohan, Taylor Wood, Fahad Khan, Ripple Khera, Jean Michel Rouly, Anshu Jain, Jeff Sherman and Emily Vucic

Zephyr AI 1800 Tysons Blvd Suite 901, McLean VA 22102 | www.zephyrai.bio

Abstract # 4640

Bridging the Predictive Biomarker Gap: AI-Powered Drug Response Prediction Using Clinicogenomics Data Available in Real-World Settings

- Precision oncology remains limited by the lack of effective biomarkers. Few tumors harbor actionable alterations, and even when present, many patients fail to benefit. Conversely, some patients without recognized biomarkers respond to therapies—highlighting the need for more robust, biologically informed approaches.
- Traditional biomarkers, based on single mutations or genomic signatures, often miss critical drivers of drug response—especially in late-line, heterogeneous populations.
- AIM-Bx, Zephyr AI's multi-modal, AI-enabled software-based platform, addresses this gap by integrating clinical, molecular, pharmacological, and phenotypic data to generate real-world validated drug response predictions.
- The platform uses data routinely available in clinical settings—including standard features and genomic inputs from most commercial NGS and LDT panels—supporting both retrospective validation and prospective deployment.
- As a use case, AIM-Bx predicted osimertinib sensitivity in NSCLC using tissue and liquid biopsy data, identifying EGFR+ non-responders and EGFR– responders. These results highlight AIM-Bx's ability to uncover hidden biological drivers, expand targeted therapy utility, and enable longitudinal prediction in real-world contexts.

Fig 1 | AI-Enabled Multi-Modal Biomarkers (AIM-Bx) Architecture Overview.

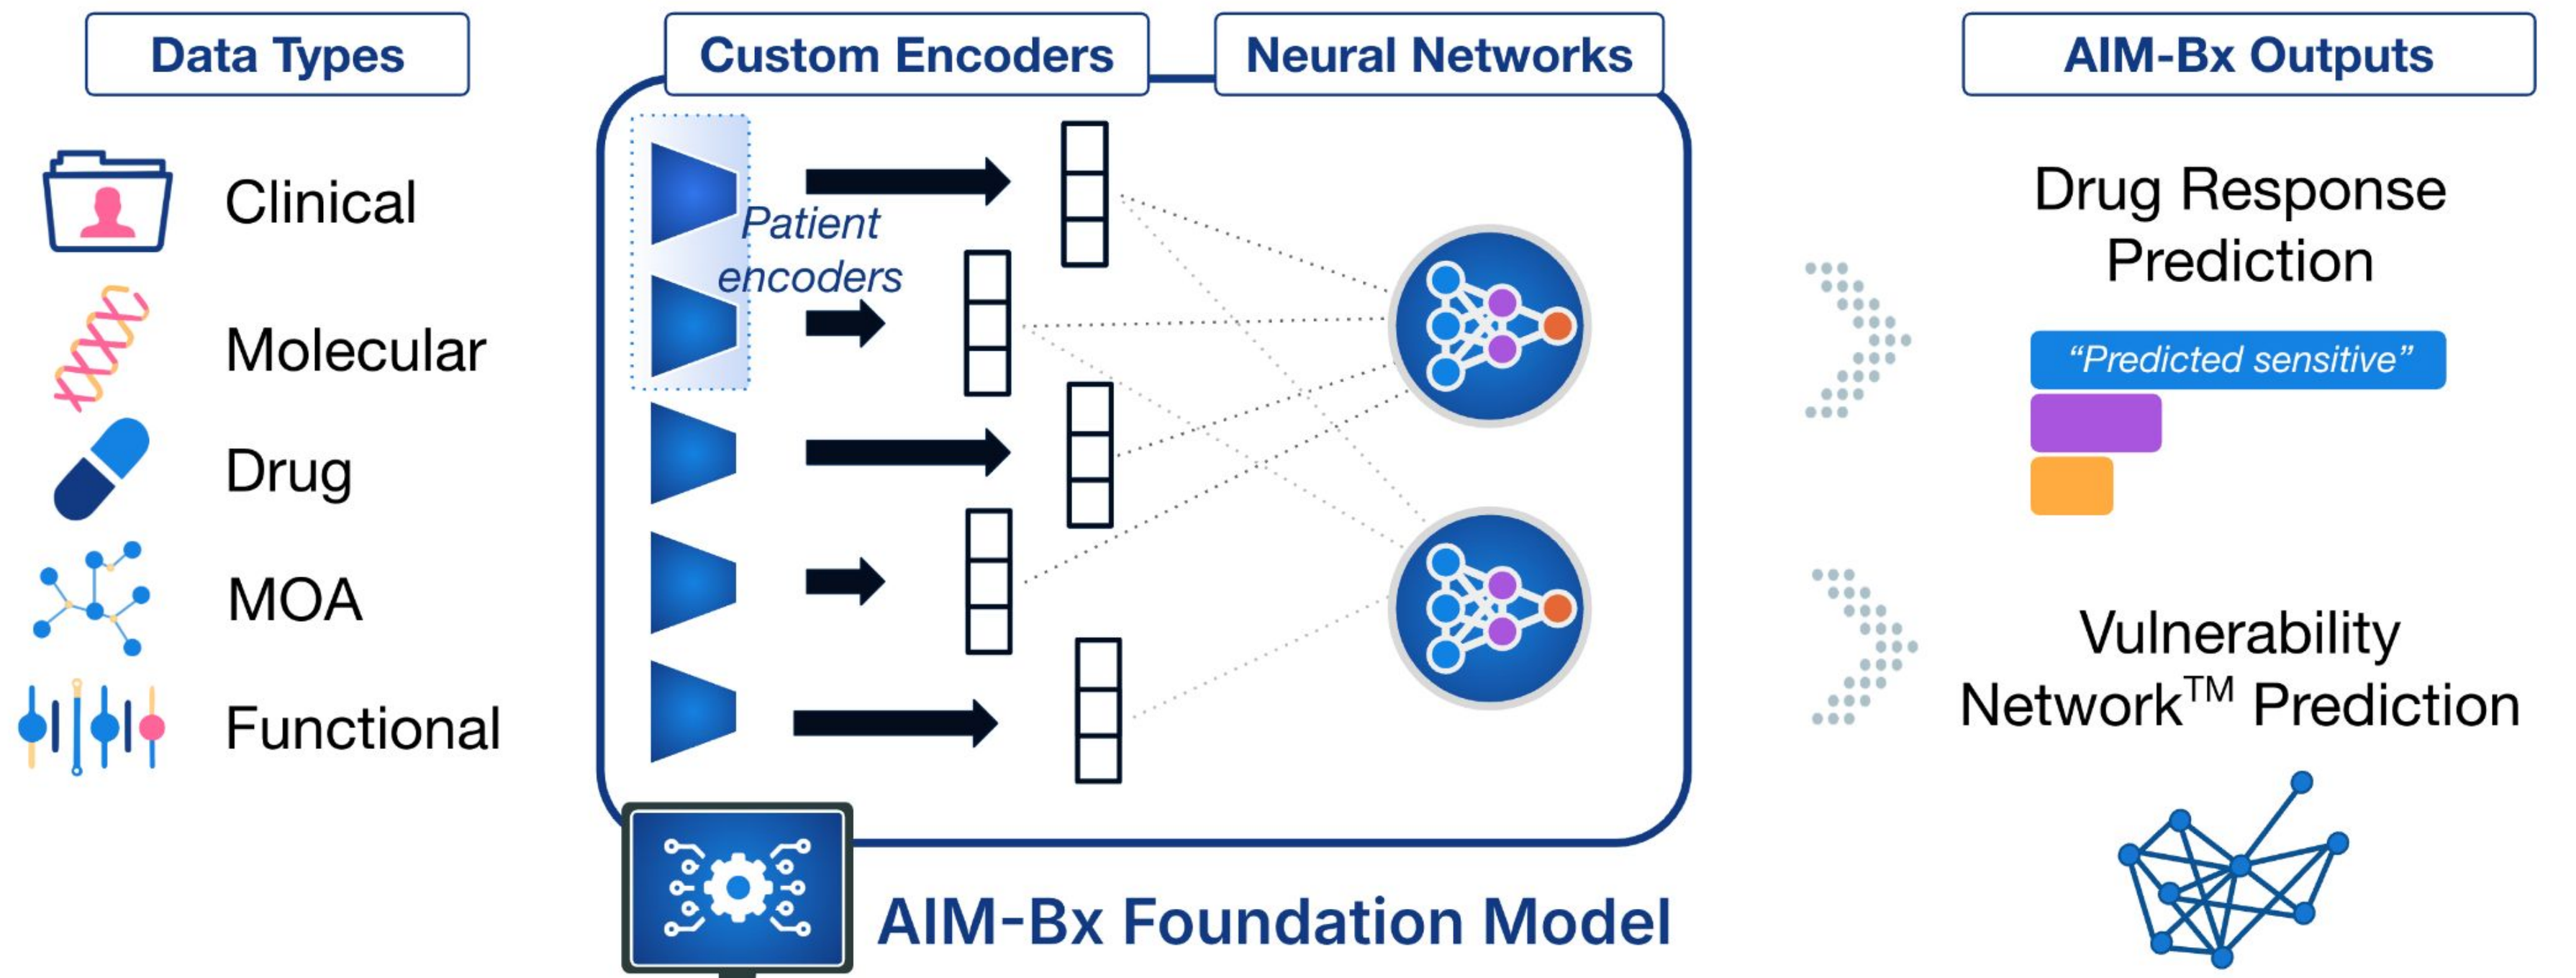
Data Types
Clinical
Molecular
Drug
MOA
Functional
Custom Encoders
Neural Networks
Patient encoders
AIM-Bx Foundation Model
AIM-Bx Outputs
Drug Response Prediction
"Predicted sensitive"
Vulnerability Network™ Prediction

The AIM-Bx Foundation Model encodes complex data modalities—including patient-level clinicogenomic features and drug structure—into rich, ML-ready representations. These are processed by neural networks trained to predict drug response and generate Vulnerability Networks™ that reveal predicted tumor dependencies relevant to response.

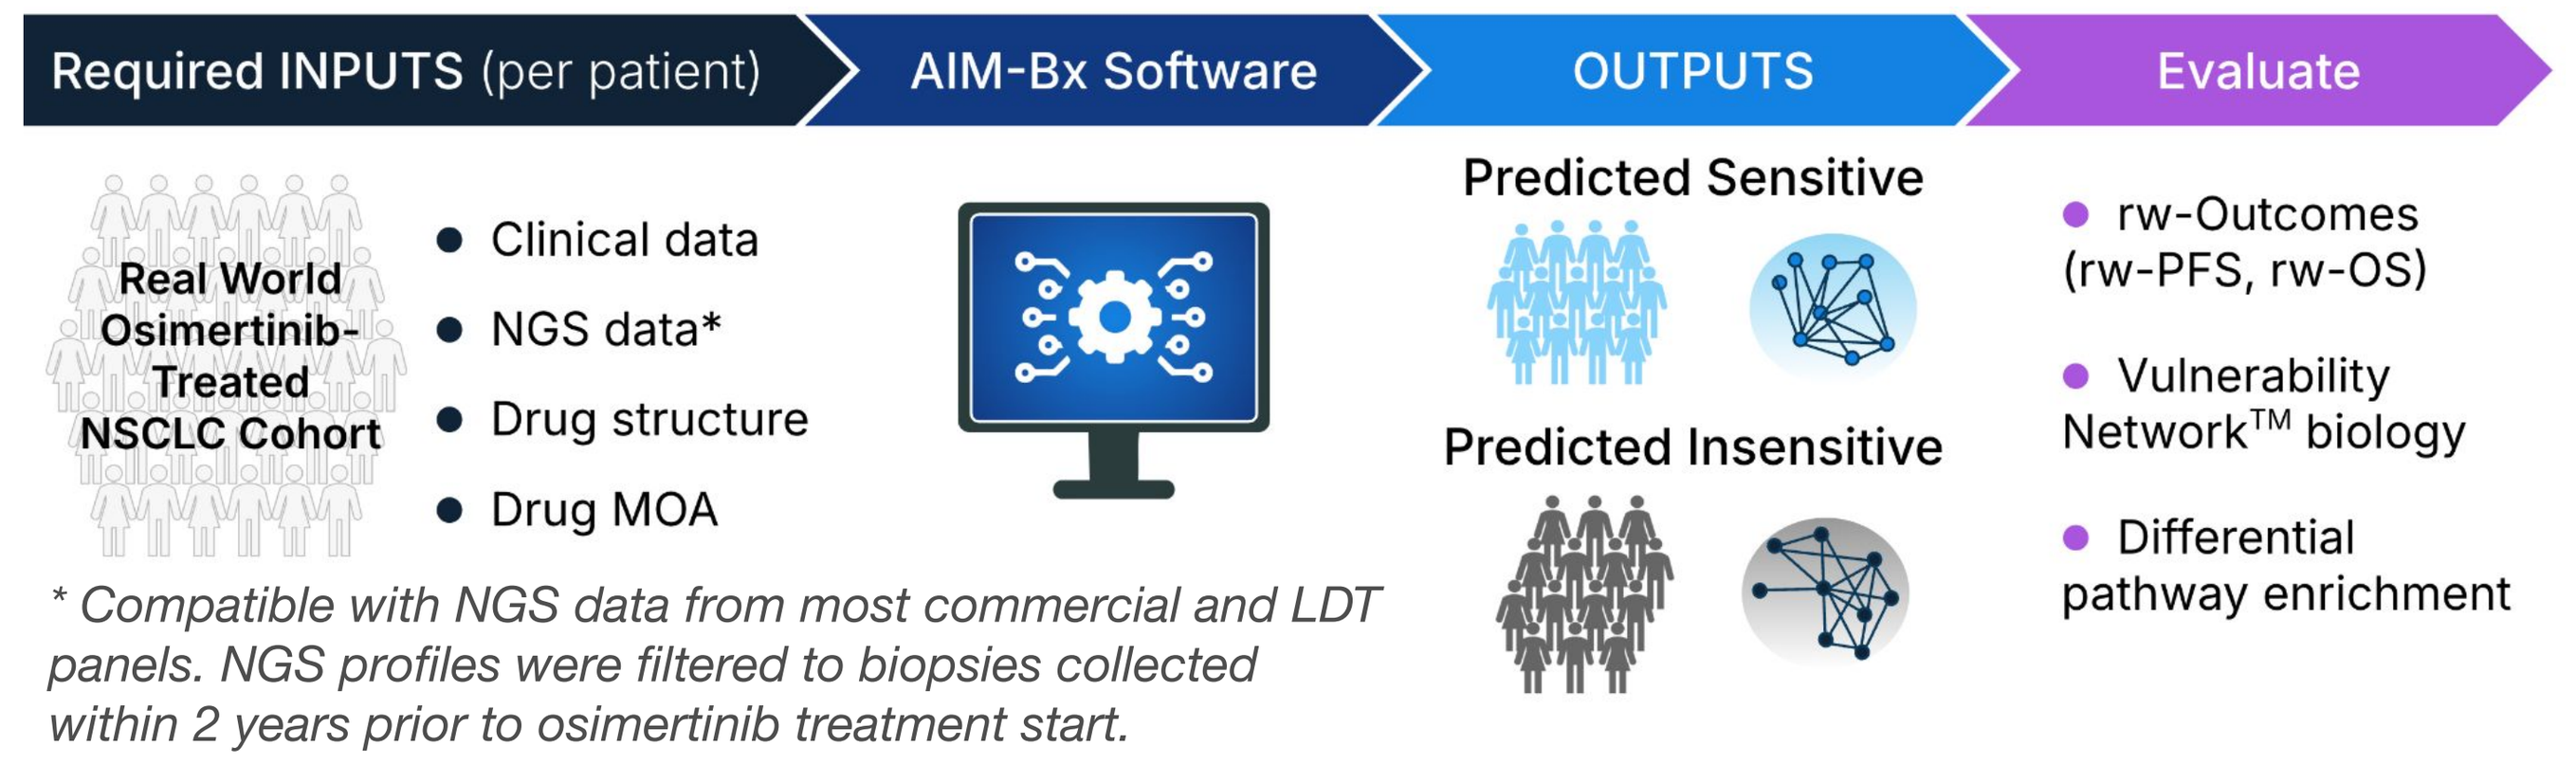
Required INPUTS (per patient)
AIM-Bx Software
OUTPUTS
Evaluate
Real World Osimertinib-Treated NSCLC Cohort
Clinical data
NGS data*
Drug structure
Drug MOA
Predicted Sensitive
Predicted Insensitive
rw-Outcomes (rw-PFS, rw-OS)
Vulnerability Network™ biology
Differential pathway enrichment
* Compatible with NGS data from most commercial and LDT panels. NGS profiles were filtered to biopsies collected within 2 years prior to osimertinib treatment start.

Fig 2 | Evaluation Framework.

Clinical & NGS data to run AIM-Bx predictions, along with real-world treatment outcomes for post-hoc evaluation, were sourced from public (AACR Project GENIE) and non-public data (CancerLinQ, Aster Insights™ Optum® Market Clarity, Guardant Health™).

Sensitive and non-sensitive groups were biologically characterized post hoc using Vulnerability Networks™ and gene set enrichment analysis of reconstructed gene expression from the AIM-Bx patient encoder (Fig 1).

AIM-Bx Stratifies NSCLC Patients by Osimertinib Sensitivity with Clinically Meaningful Outcomes Across Diverse Real-World Cohorts

Fig 3 | AIM-Bx Identifies Osimertinib Responders Independent of EGFR Status.

NSCLC clinicogenomic and outcome data were sourced from AACR Project GENIE (N=97), CancerLinQ (N=32), and Aster Insights (N=21). **[A]** AIM-Bx–predicted osimertinib-sensitive NSCLC patients had significantly improved rw-OS compared to predicted insensitive patients (median 10 months; HR = 0.54 indexed from treatment start). **[B]** Most patients were on-label (EGFR+) and AIM-Bx predictions (ZephyrAI) were significantly associated with survival in a univariable Cox PH analysis adjusting for available confounders. **[C]** EGFR variants were not significantly enriched in AIM-Bx–predicted sensitive vs insensitive groups.

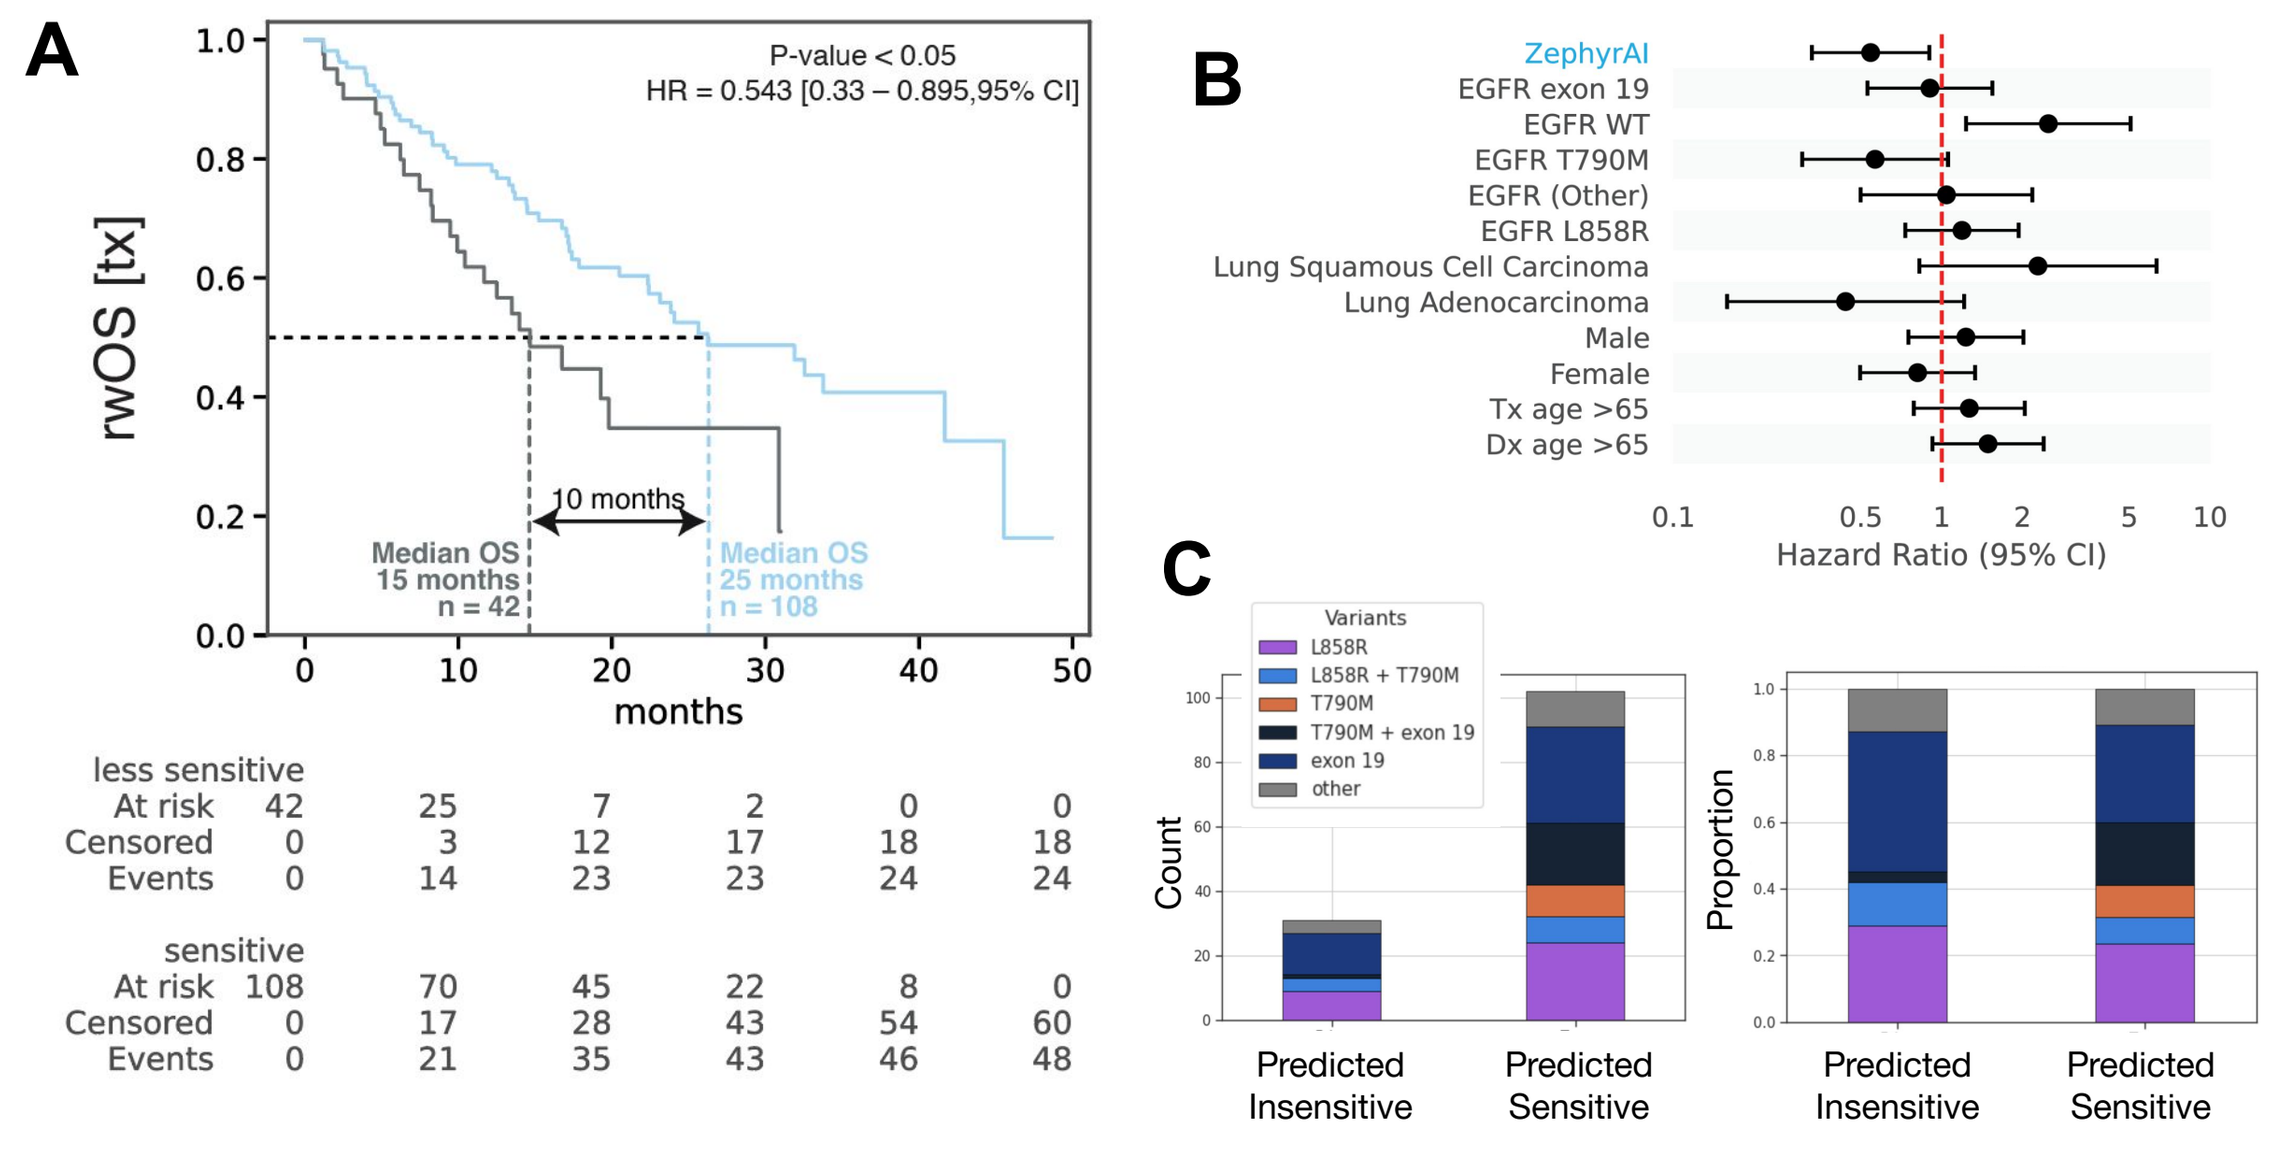
A
P-value < 0.05
HR = 0.543 [0.33 – 0.895,95% CI]
rwOS [tx]
months
10 months
Median OS 15 months n = 42
Median OS 25 months n = 108
less sensitive
sensitive
At risk
Censored
Events
B
ZephyrAI
EGFR exon 19
EGFR WT
EGFR T790M
EGFR (Other)
EGFR L858R
Lung Squamous Cell Carcinoma
Lung Adenocarcinoma
Male
Female
Tx age >65
Dx age >65
Hazard Ratio (95% CI)
C
Variants
L858R
L858R + T790M
T790M
T790M + exon 19
exon 19
other
Count
Proportion
Predicted Insensitive
Predicted Sensitive

Fig 4 | AIM-Bx Predicts Improved Real-World Outcomes in an Expanded rw-NSCLC Cohort.

An expanded NSCLC cohort (N=308) from Optum® Market Clarity† was analyzed in a secure ODDW environment. AIM-Bx–predicted sensitive patients had significantly improved outcomes compared to non-sensitive patients: **[A]** rw-PFS: +16 months median survival (HR = 0.51, $p < 0.001$) and **[B]** rw-OS: +17 months median survival (HR = 0.50, $p < 0.001$). Real-world outcomes were indexed to osimertinib treatment start. AIM-Bx predictions (ZephyrAI) were the only factor significantly associated with improved survival in univariable Cox PH models adjusting for available confounders. Results support AIM-Bx performance across diverse real-world settings and heterogeneous NGS inputs.

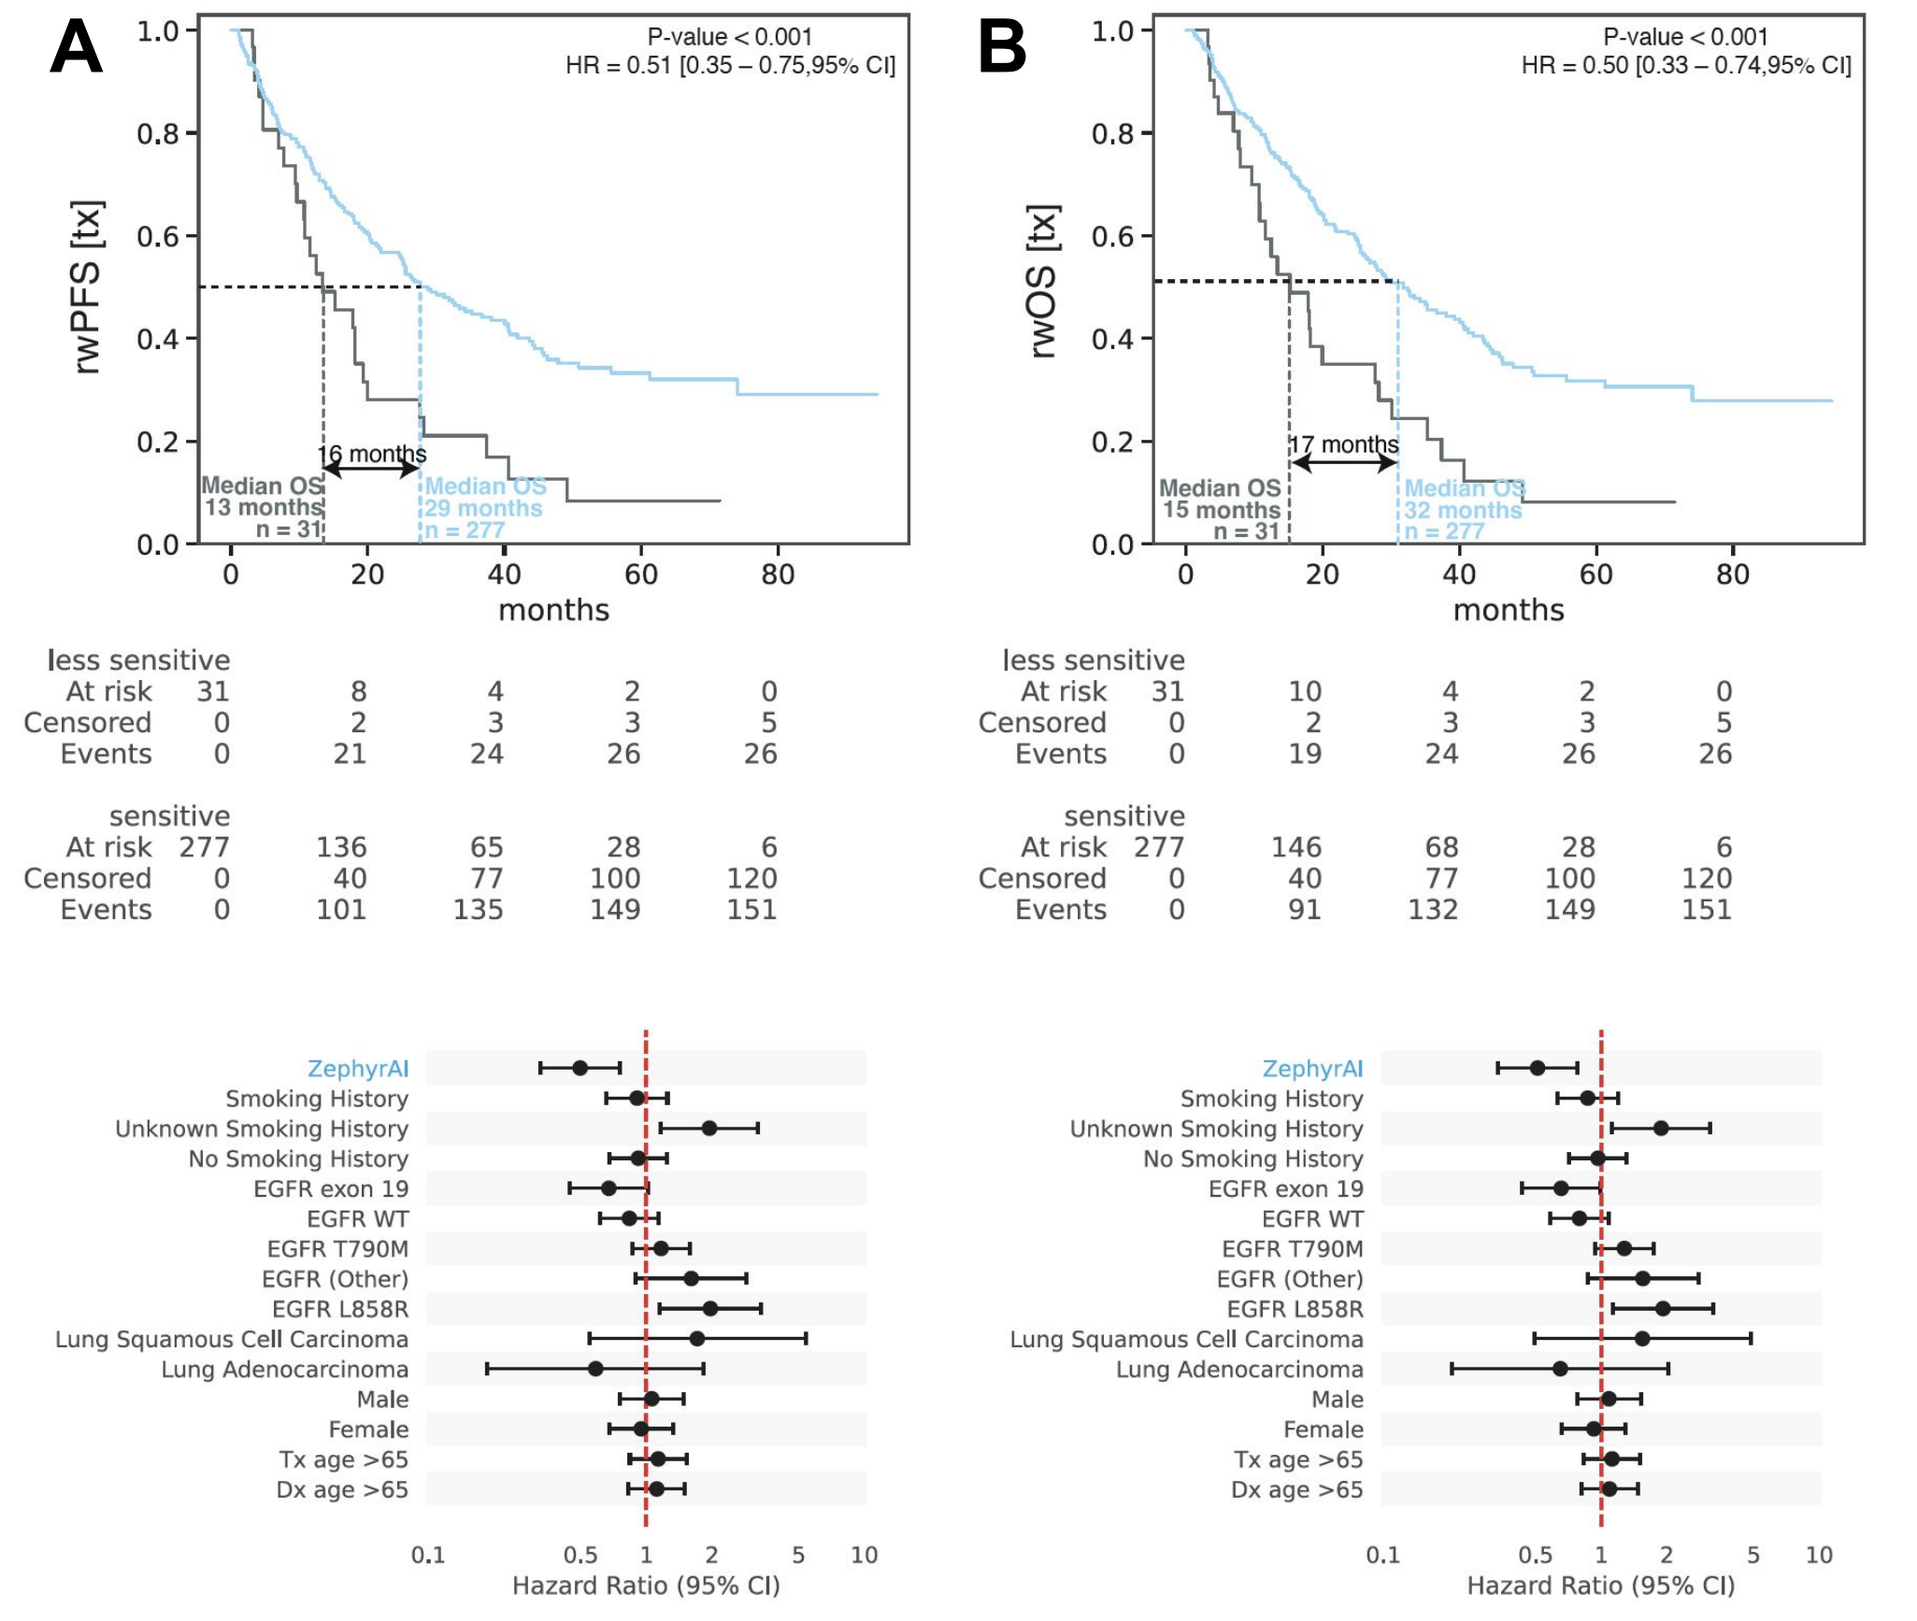
A
P-value < 0.001
HR = 0.51 [0.35 – 0.75,95% CI]
rwPFS [tx]
16 months
Median OS 13 months n = 31
Median OS 29 months n = 277
B
P-value < 0.001
HR = 0.50 [0.33 – 0.74,95% CI]
rwOS [tx]
17 months
Median OS 15 months n = 31
Median OS 32 months n = 277
months
less sensitive
sensitive
At risk
Censored
Events
ZephyrAI
Smoking History
Unknown Smoking History
No Smoking History
EGFR exon 19
EGFR WT
EGFR T790M
EGFR (Other)
EGFR L858R
Lung Squamous Cell Carcinoma
Lung Adenocarcinoma
Male
Female
Tx age >65
Dx age >65
Hazard Ratio (95% CI)

Fig 5 | AIM-Bx Validates Osimertinib Predictions Using Liquid Biopsy–Derived NGS Data.

A NSCLC cohort (N=435) treated with osimertinib and profiled via ctDNA (Guardant Health™) was used to evaluate AIM-Bx on liquid biopsy inputs. AIM-Bx–predicted sensitive patients had significantly improved outcomes: **[A]** rw-OS: +18 months; HR = 0.70, $p < 0.05$ and **[B]** rw-PFS: +20 months; HR = 0.72, $p < 0.05$, indexed to osimertinib treatment start dates. AIM-Bx predictions are significantly associated with improved survival in univariable Cox models, alongside patient sex (female) as an independent factor. Results support AIM-Bx performance across diverse NGS inputs, including ctDNA, positioning AIM-Bx for broader clinical integration and longitudinal modeling.

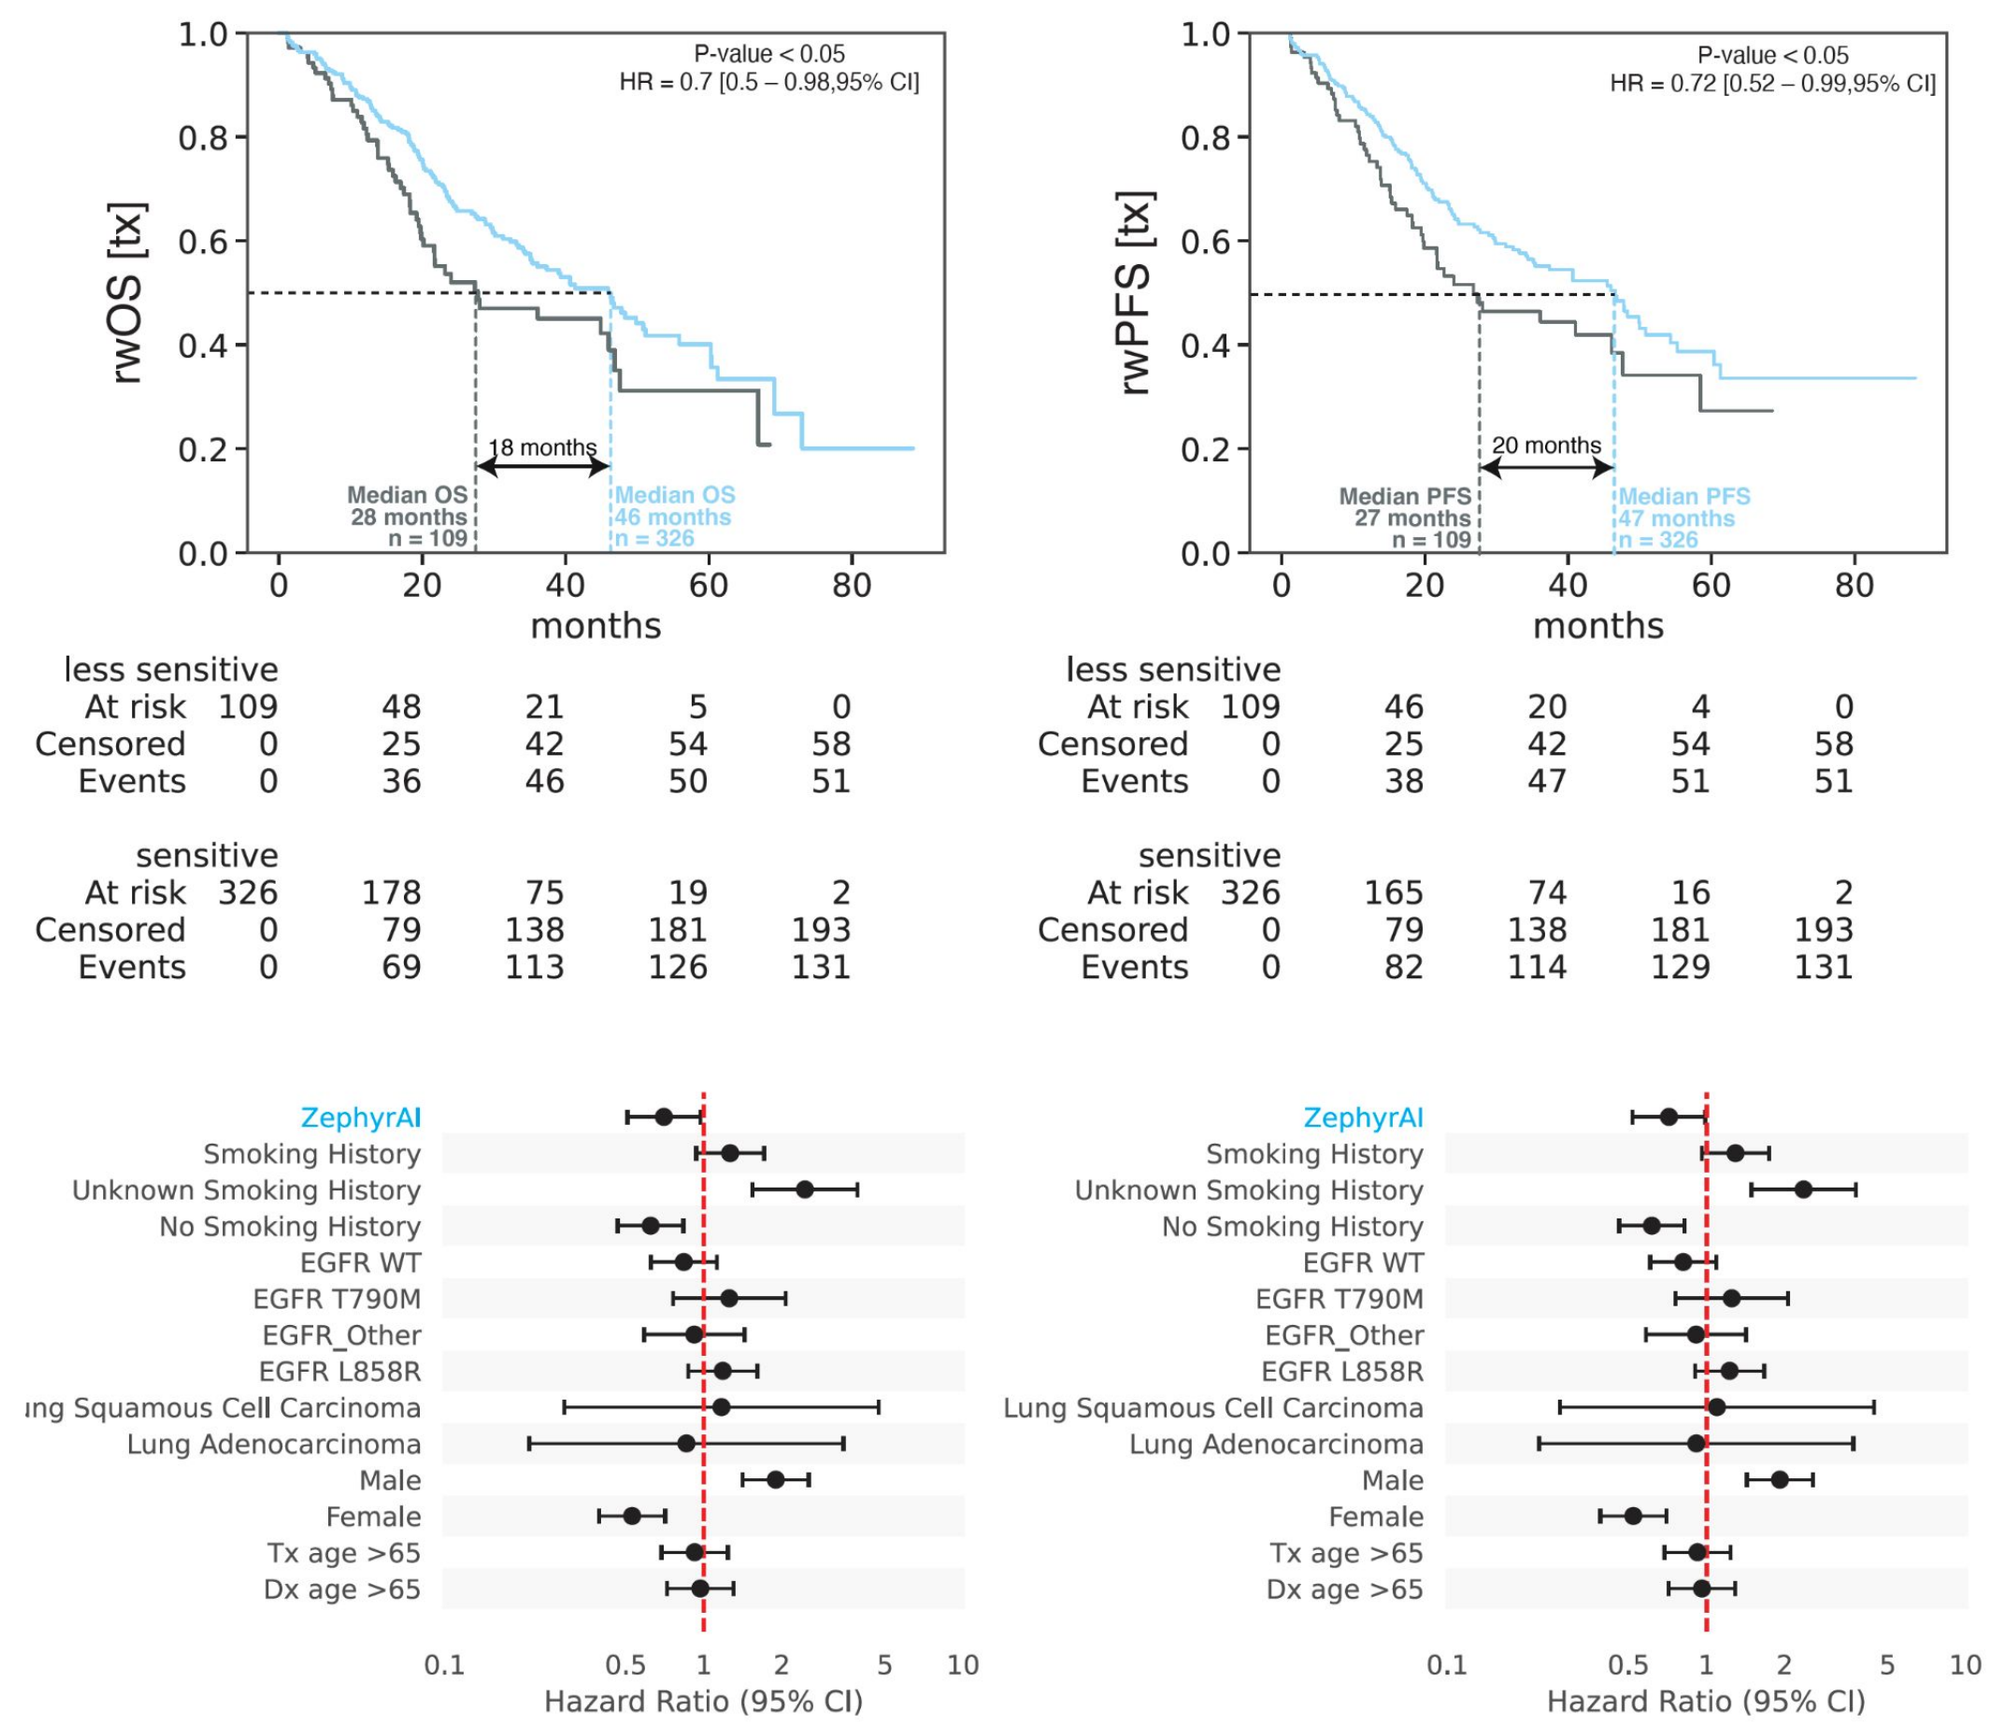
P-value < 0.05
HR = 0.7 [0.5 – 0.98,95% CI]
rwOS [tx]
18 months
Median OS 28 months n = 109
Median OS 46 months n = 326
P-value < 0.05
HR = 0.72 [0.52 – 0.99,95% CI]
rwPFS [tx]
20 months
Median PFS 27 months n = 109
Median PFS 47 months n = 326
months
less sensitive
sensitive
At risk
Censored
Events
ZephyrAI
Smoking History
Unknown Smoking History
No Smoking History
EGFR WT
EGFR T790M
EGFR_Other
EGFR L858R
Lung Squamous Cell Carcinoma
Lung Adenocarcinoma
Male
Female
Tx age >65
Dx age >65
Hazard Ratio (95% CI)

Uncovering Tumor Dependencies and Transcriptomic Programs Driving Osimertinib Response via VNs and Reconstructed mRNA

Fig 6 | Vulnerability Networks Reveal Distinct Tumor Dependencies Associated with Osimertinib Response Not Captured by EGFR Genomic Status.

VNs were generated for NSCLC tumors from Fig. 3, with two networks significantly enriched in each AIM-Bx–predicted sensitive and insensitive groups. VNs reveal predicted patient-specific mechanisms underlying AIM-Bx response predictions.

[A] Responder VNs were characterized by dependencies on EGFR, EGFR signaling and key survival and metabolic pathways, suggesting that network-level activity—beyond EGFR DNA level alterations— drive sensitivity to osimertinib.

[B] Non-responding NSCLC VNs were characterized by dependencies in cell cycle, transcriptional, and integrin signaling pathways, consistent with EGFR independence and known osimertinib resistance mechanisms.

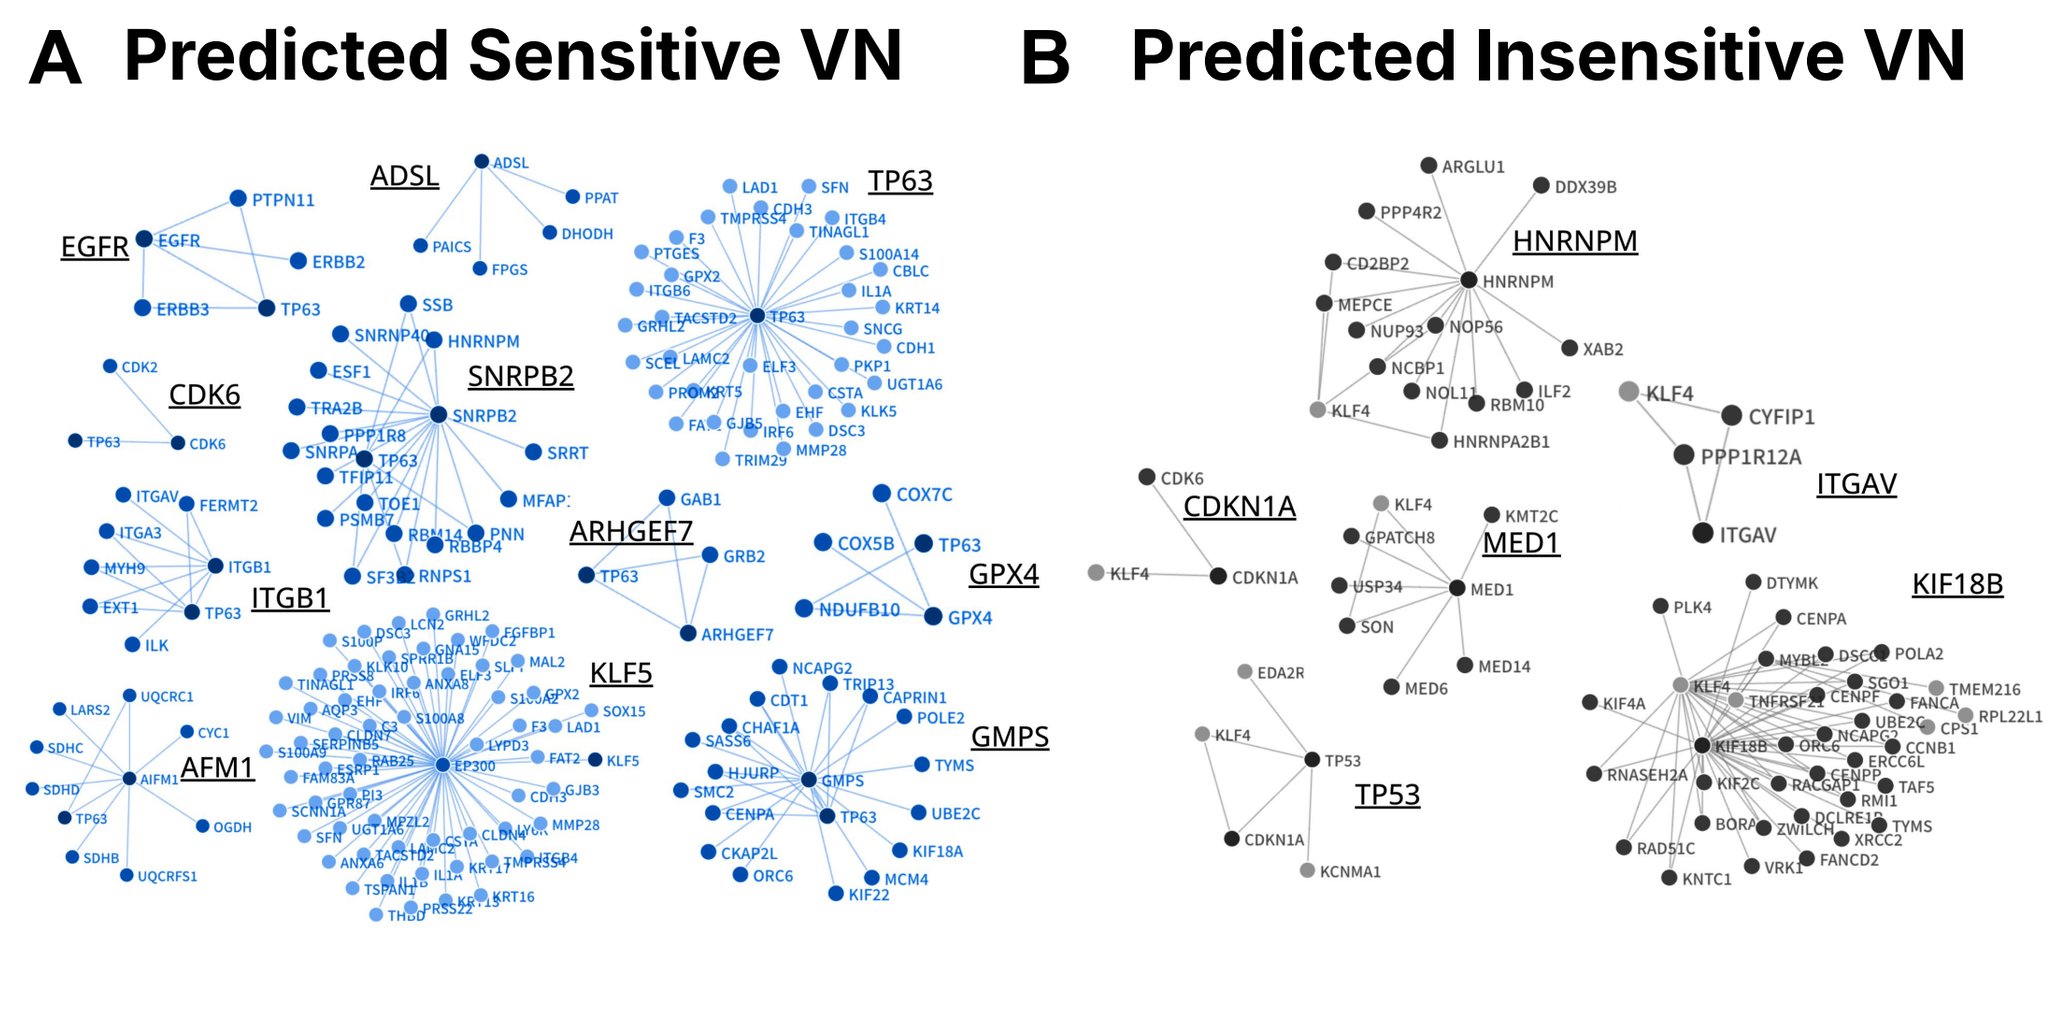
A Predicted Sensitive VN
B Predicted Insensitive VN
EGFR
ADSL
TP63
CDK6
SNRPB2
ITGB1
ARHGEF7
GPX4
AFM1
KLF5
GMPS
HNRNPM
CDKN1A
MED1
ITGAV
KIF18B
TP53

Fig 7 | GSEA of Reconstructed mRNA Profiles Reveals Distinct Biological Programs Across AIM-Bx Predicted Response Groups.

Reconstructed mRNA profiles for tumors in Fig. 3 were generated using the AIM-Bx Patient Encoder (Fig. 1) from clinical and genomic inputs, and analyzed by GSEA.

Less-sensitive tumors were enriched for oncogenic programs linked to proliferation and poor outcomes in NSCLC, including DREAM, E2F, MYC, KRAS, and NRF2—pathways previously associated with TKI resistance, including resistance to osimertinib.

In contrast, predicted osimertinib-sensitive tumors were enriched for EGFR signaling, kinase inhibitor response signatures (e.g., gefitinib, dasatinib) and deactivation of stem-like programs suggesting a less proliferative, TKI-responsive phenotype.

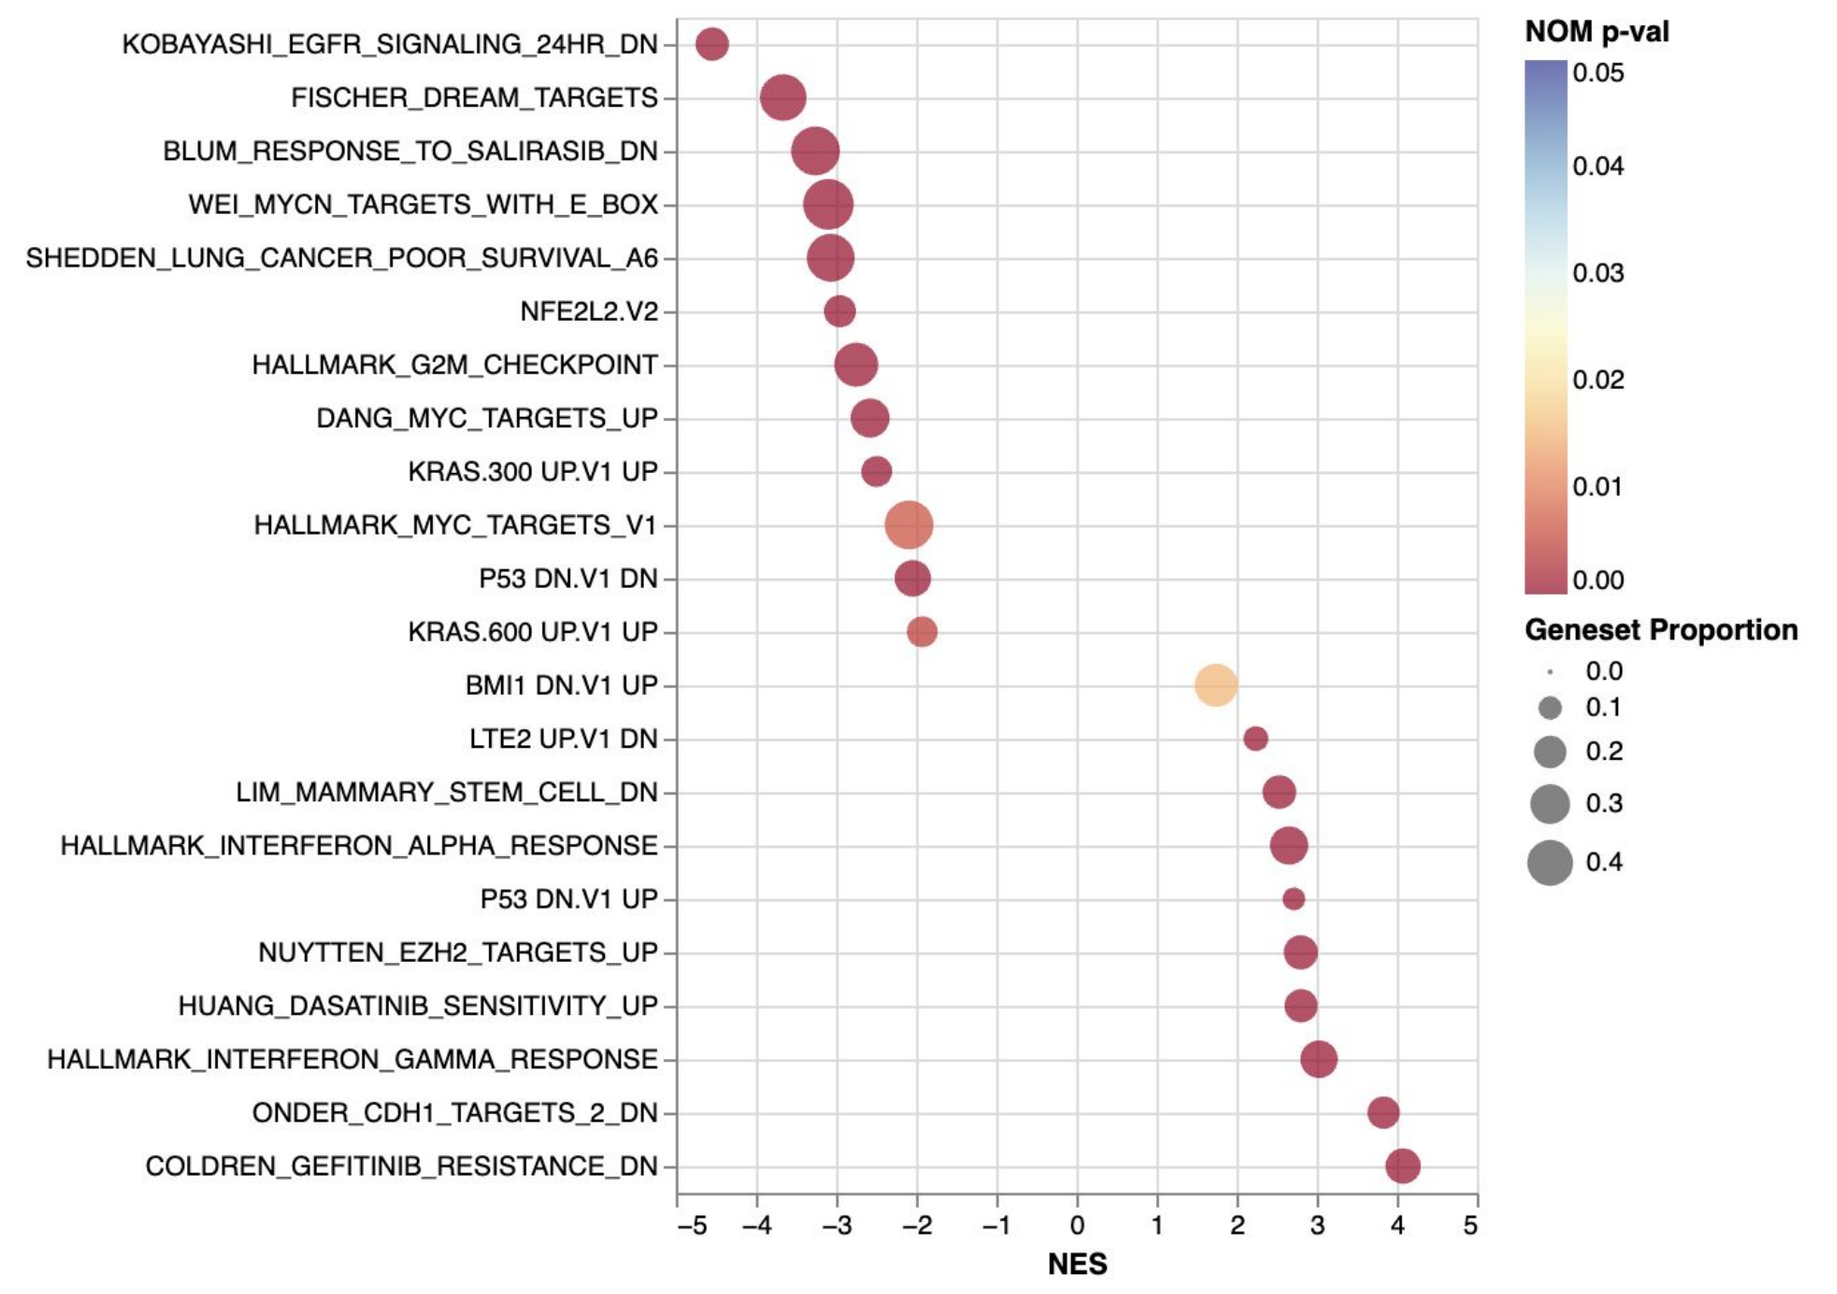
KOBAYASHI_EGFR_SIGNALING_24HR_DN
FISCHER_DREAM_TARGETS
BLUM_RESPONSE_TO_SALIRASIB_DN
WEI_MYCN_TARGETS_WITH_E_BOX
SHEDDEN_LUNG_CANCER_POOR_SURVIVAL_A6
NFE2L2.V2
HALLMARK_G2M_CHECKPOINT
DANG_MYC_TARGETS_UP
KRAS.300 UP.V1 UP
HALLMARK_MYC_TARGETS_V1
P53 DN.V1 DN
KRAS.600 UP.V1 UP
BMI1 DN.V1 UP
LTE2 UP.V1 DN
LIM_MAMMARY_STEM_CELL_DN
HALLMARK_INTERFERON_ALPHA_RESPONSE
P53 DN.V1 UP
NUYTTEN_EZH2_TARGETS_UP
HUANG_DASATINIB_SENSITIVITY_UP
HALLMARK_INTERFERON_GAMMA_RESPONSE
ONDER_CDH1_TARGETS_2_DN
COLDREN_GEFITINIB_RESISTANCE_DN
NES
NOM p-val
Geneset Proportion

Normalized enrichment scores (NES, x-axis) reflect gene set enrichment (y-axis) from MSigDB collections (c2, hallmark, oncogenic). Positive NES values indicate enrichment in AIM-Bx–predicted sensitive tumors; negative NES in non-sensitive tumors. Circle size reflects gene set size; color indicates statistical significance (p-value). Differential expression was performed with Limma and filtered for p-value < 0.05.

Platform Summary & Future Directions

- AIM-Bx predicts drug response using routinely available clinical and genomic data from tissue or liquid biopsy, revealing tumor vulnerabilities and biological programs beyond traditional DNA biomarkers.
- Validated using osimertinib as a proof-of-concept, AIM-Bx is adaptable to novel drugs, mechanisms, and indications through scalable, transfer learning–based model development.
- Our software seamlessly integrates into clinical workflows and supports rapid deployment in trials or retrospective studies using standard NGS and LDT panel data.
- Zephyr AI welcomes partnerships to co-develop AI-enabled CDx strategies for precision oncology.

† Optum's de-identified Market Clarity Data (Optum® Market Clarity) is an integrated, multi-source medical claims, pharmacy claims, and electronic health records data set. Optum® Market Clarity links electronic health record data - including lab results, vital signs and measurements, diagnoses, procedures and information derived from unstructured clinical notes using natural language processing - with historical, linked administrative claim data - including pharmacy claims, physician claims, clinical information facility claims and medications prescribed and administered. Optum® Market Clarity is statistically de-identified under the HIPAA Privacy Rule's Expert Determination method and managed according to Optum® customer data use agreements.

Contact: partnerships@zephyrai.bio